DISCOVERY OF POTENT AND SELECTIVE EP300 DEGRADERS WITH ANTI-CANCER ACTIVITY

Mark W. Zimmerman[1], Ammar Adam[1], Hafiz, Ahmad[1], Benjamin Adams[1], Ketaki Adhikari[1], Wesley Austin[1], Breanna Bullock[1], Julie Di Bernardo[1], Thomas Dixon[1], Danette Daniels[1], Claudia Dominici[1], GiNell Elliot[1], Brian Ethell[1], , Anais Gervais[1], Md Imran Hossain[1], David Huang[1], Dave Lahr[1], Laura La Bonte[1], Mei Yun Lin[1], David Mayhew[1], Karolina Mizeracka[1], Solymar Negretti[1], Tyler Nguyen[1], Olga Prifti[1], Darshan Sappal[1], Shawn Schiller[1], Brenna Sherbanee[1], David Terry[1], Nihan Ucisik[1], Elizabeth Wittenborn[1], Qianhe Zhou[1]

[1] Foghorn Therapeutics, Cambridge, MA 02139

ABSTRACT

E1A binding protein (EP300) and CREB binding protein (CBP) are paralog histone acetyltransferases involved in many cellular processes via their activity as transcriptional co-activators. Dysregulation of one or both proteins has been implicated in various cancers, and functional genomic screens have demonstrated a bidirectional synthetic lethal relationship between the two genes in tumor cells. Due to the high homology between EP300 and CBP, identifying chemical matter that selectively targets EP300 or CBP has proven challenging. Here, we describe a potent, highly selective heterobifunctional degrader of EP300 with biological activity in CBP-deficient and EP300-dependent tumor cells. Anti-proliferative effects have been demonstrated in multiple cancer types, including castration-resistant prostate tumors, highlighting the essential role of EP300 in mediating oncogenic transcription required for tumor cell growth and survival. Degradation of EP300 *in vivo* attenuated androgen-driven transcription and inhibited tumor growth in VCAP (AR+) prostate tumor xenografts. Importantly, no evidence of overt toxicity or thrombocytopenia was observed at therapeutically efficacious doses. These findings indicate that selective targeting of EP300 with targeted protein degradation is a safe and effective treatment strategy for advanced tumors.

Figure 1. EP300 is a tumor-selective gene dependency

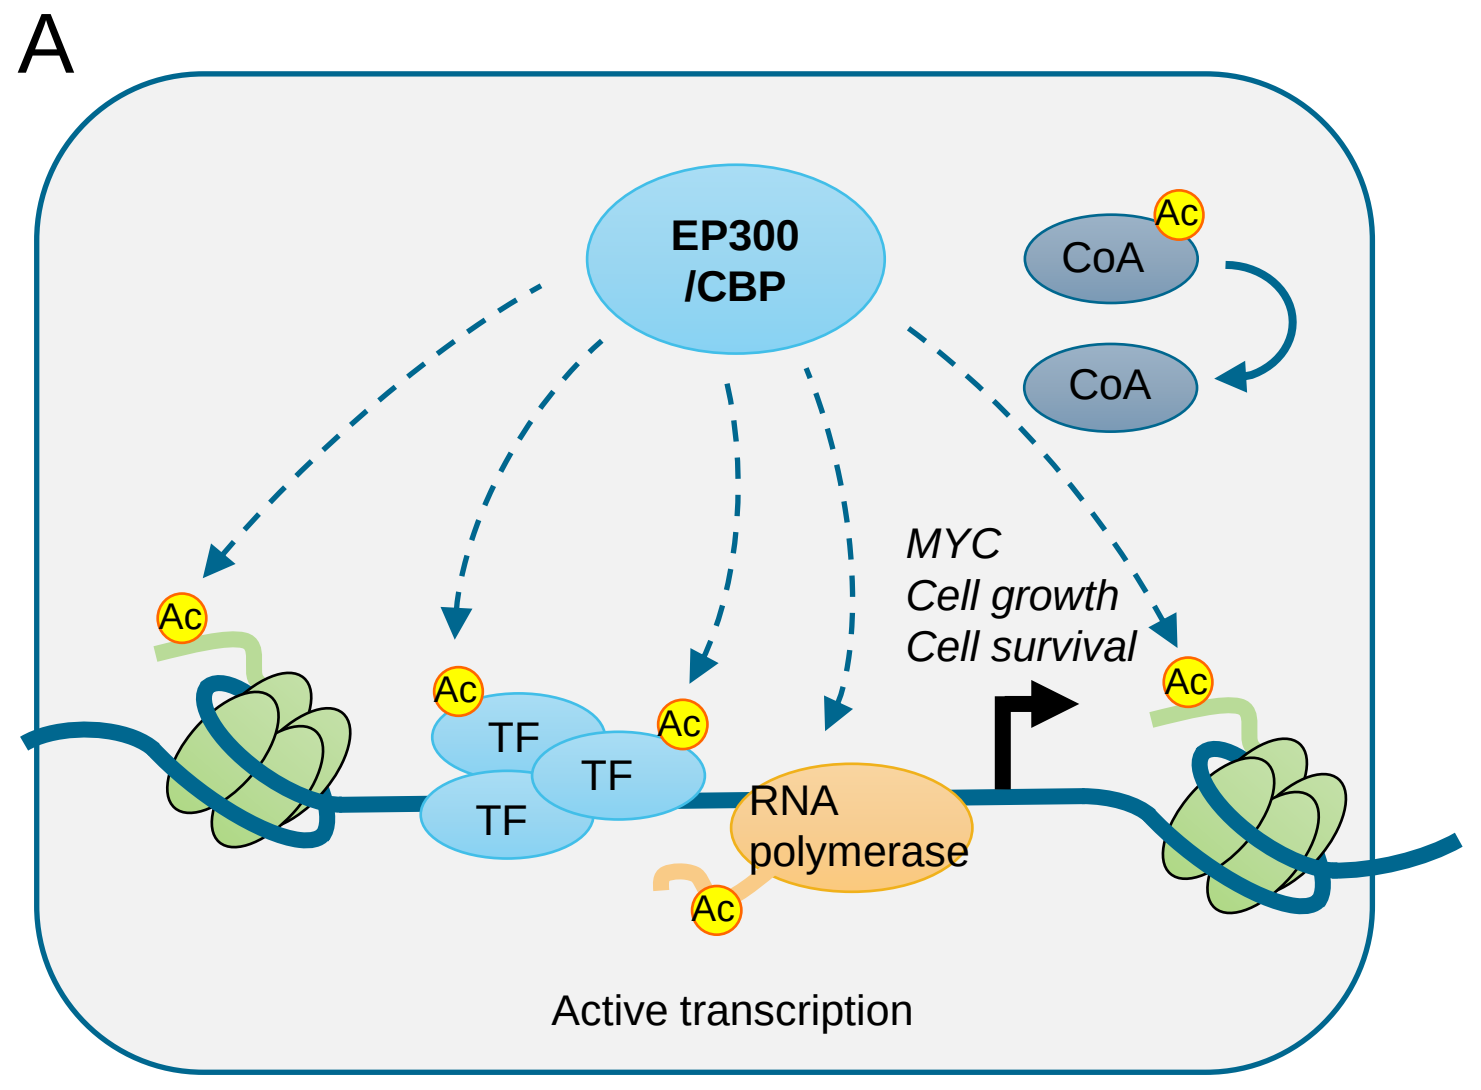
A
EP300 /CBP
CoA
Ac
MYC
Cell growth
Cell survival
TF
RNA polymerase
Active transcription

- EP300/CBP are acetyltransferases that regulate enhancer-mediated transcription and protein stability
- Quantitative acetyl-proteomics has revealed thousands of targets for EP300/CBP
- Inhibition or knockout of EP300/CBP results in downregulation of a subset of expressed genes (~10-12%)

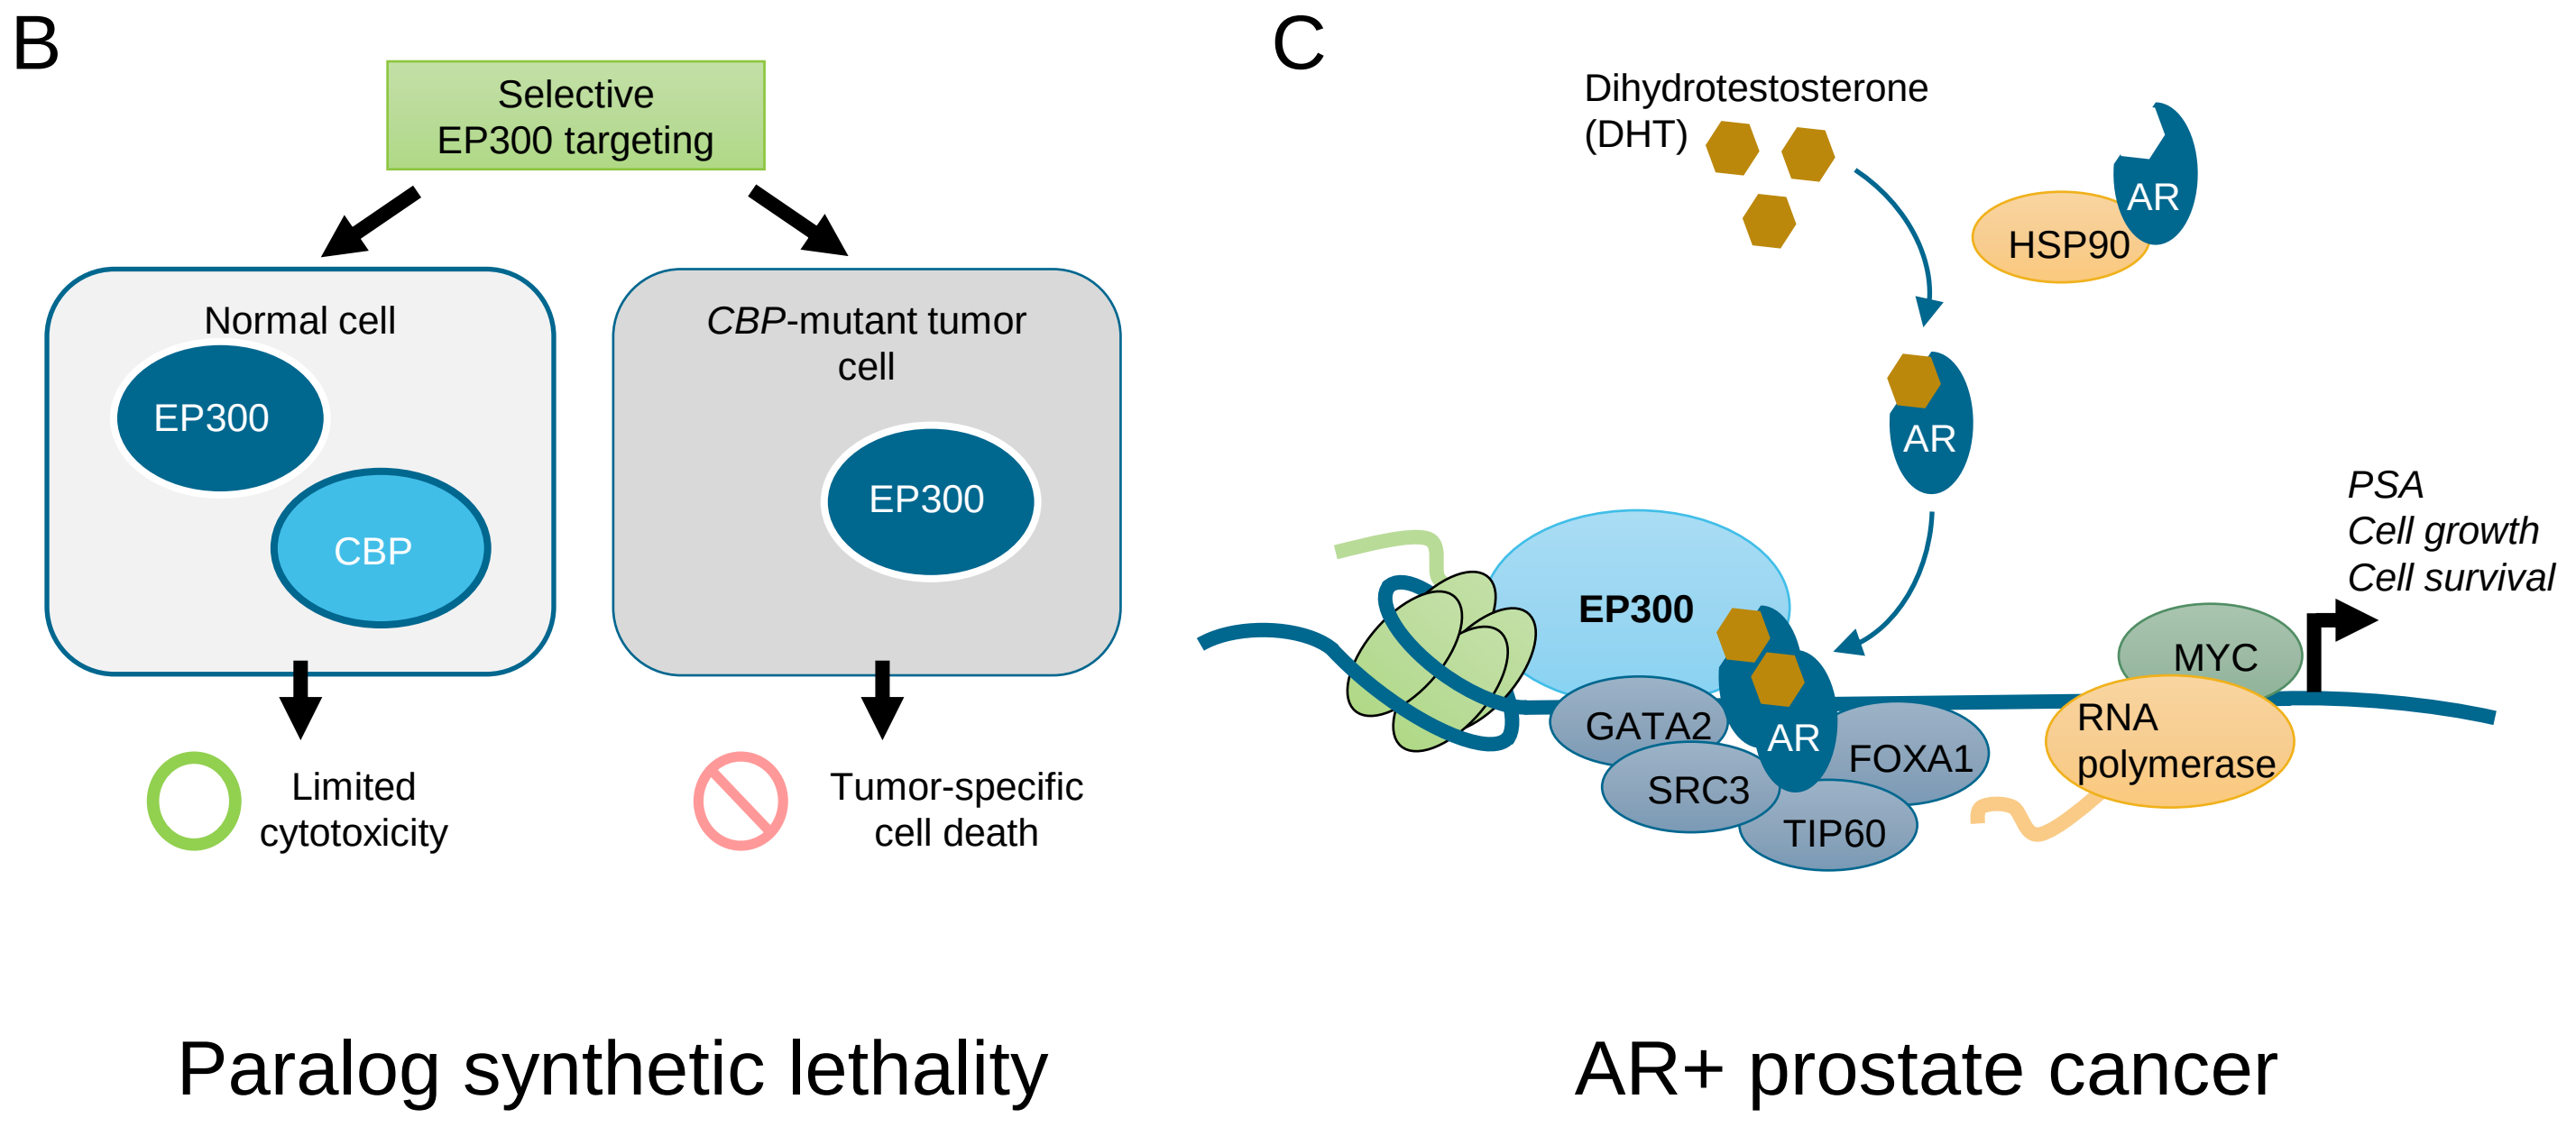
B
Selective EP300 targeting
Normal cell
EP300
CBP
CBP-mutant tumor cell
Limited cytotoxicity
Tumor-specific cell death
C
Dihydrotestosterone (DHT)
HSP90
AR
EP300
GATA2
SRC3
TIP60
FOXA1
MYC
RNA polymerase
PSA
Cell growth
Cell survival
Paralog synthetic lethality
AR+ prostate cancer

Figure 1: A) EP300 and CBP are acetyltransferase enzymes that regulate the activity and stability of a broad range of protein targets. B) CBP is frequently mutated or deleted in advanced leukemias and solid tumors, creating a synthetic lethal dependency on EP300 in tumor cells. C) EP300 is an essential co-factor for many signaling-regulated transcriptional pathways, including AR-mediated gene expression in prostate adenocarcinoma.

RESULTS

Figure 2. Identification of potent, selective EP300 Degraders

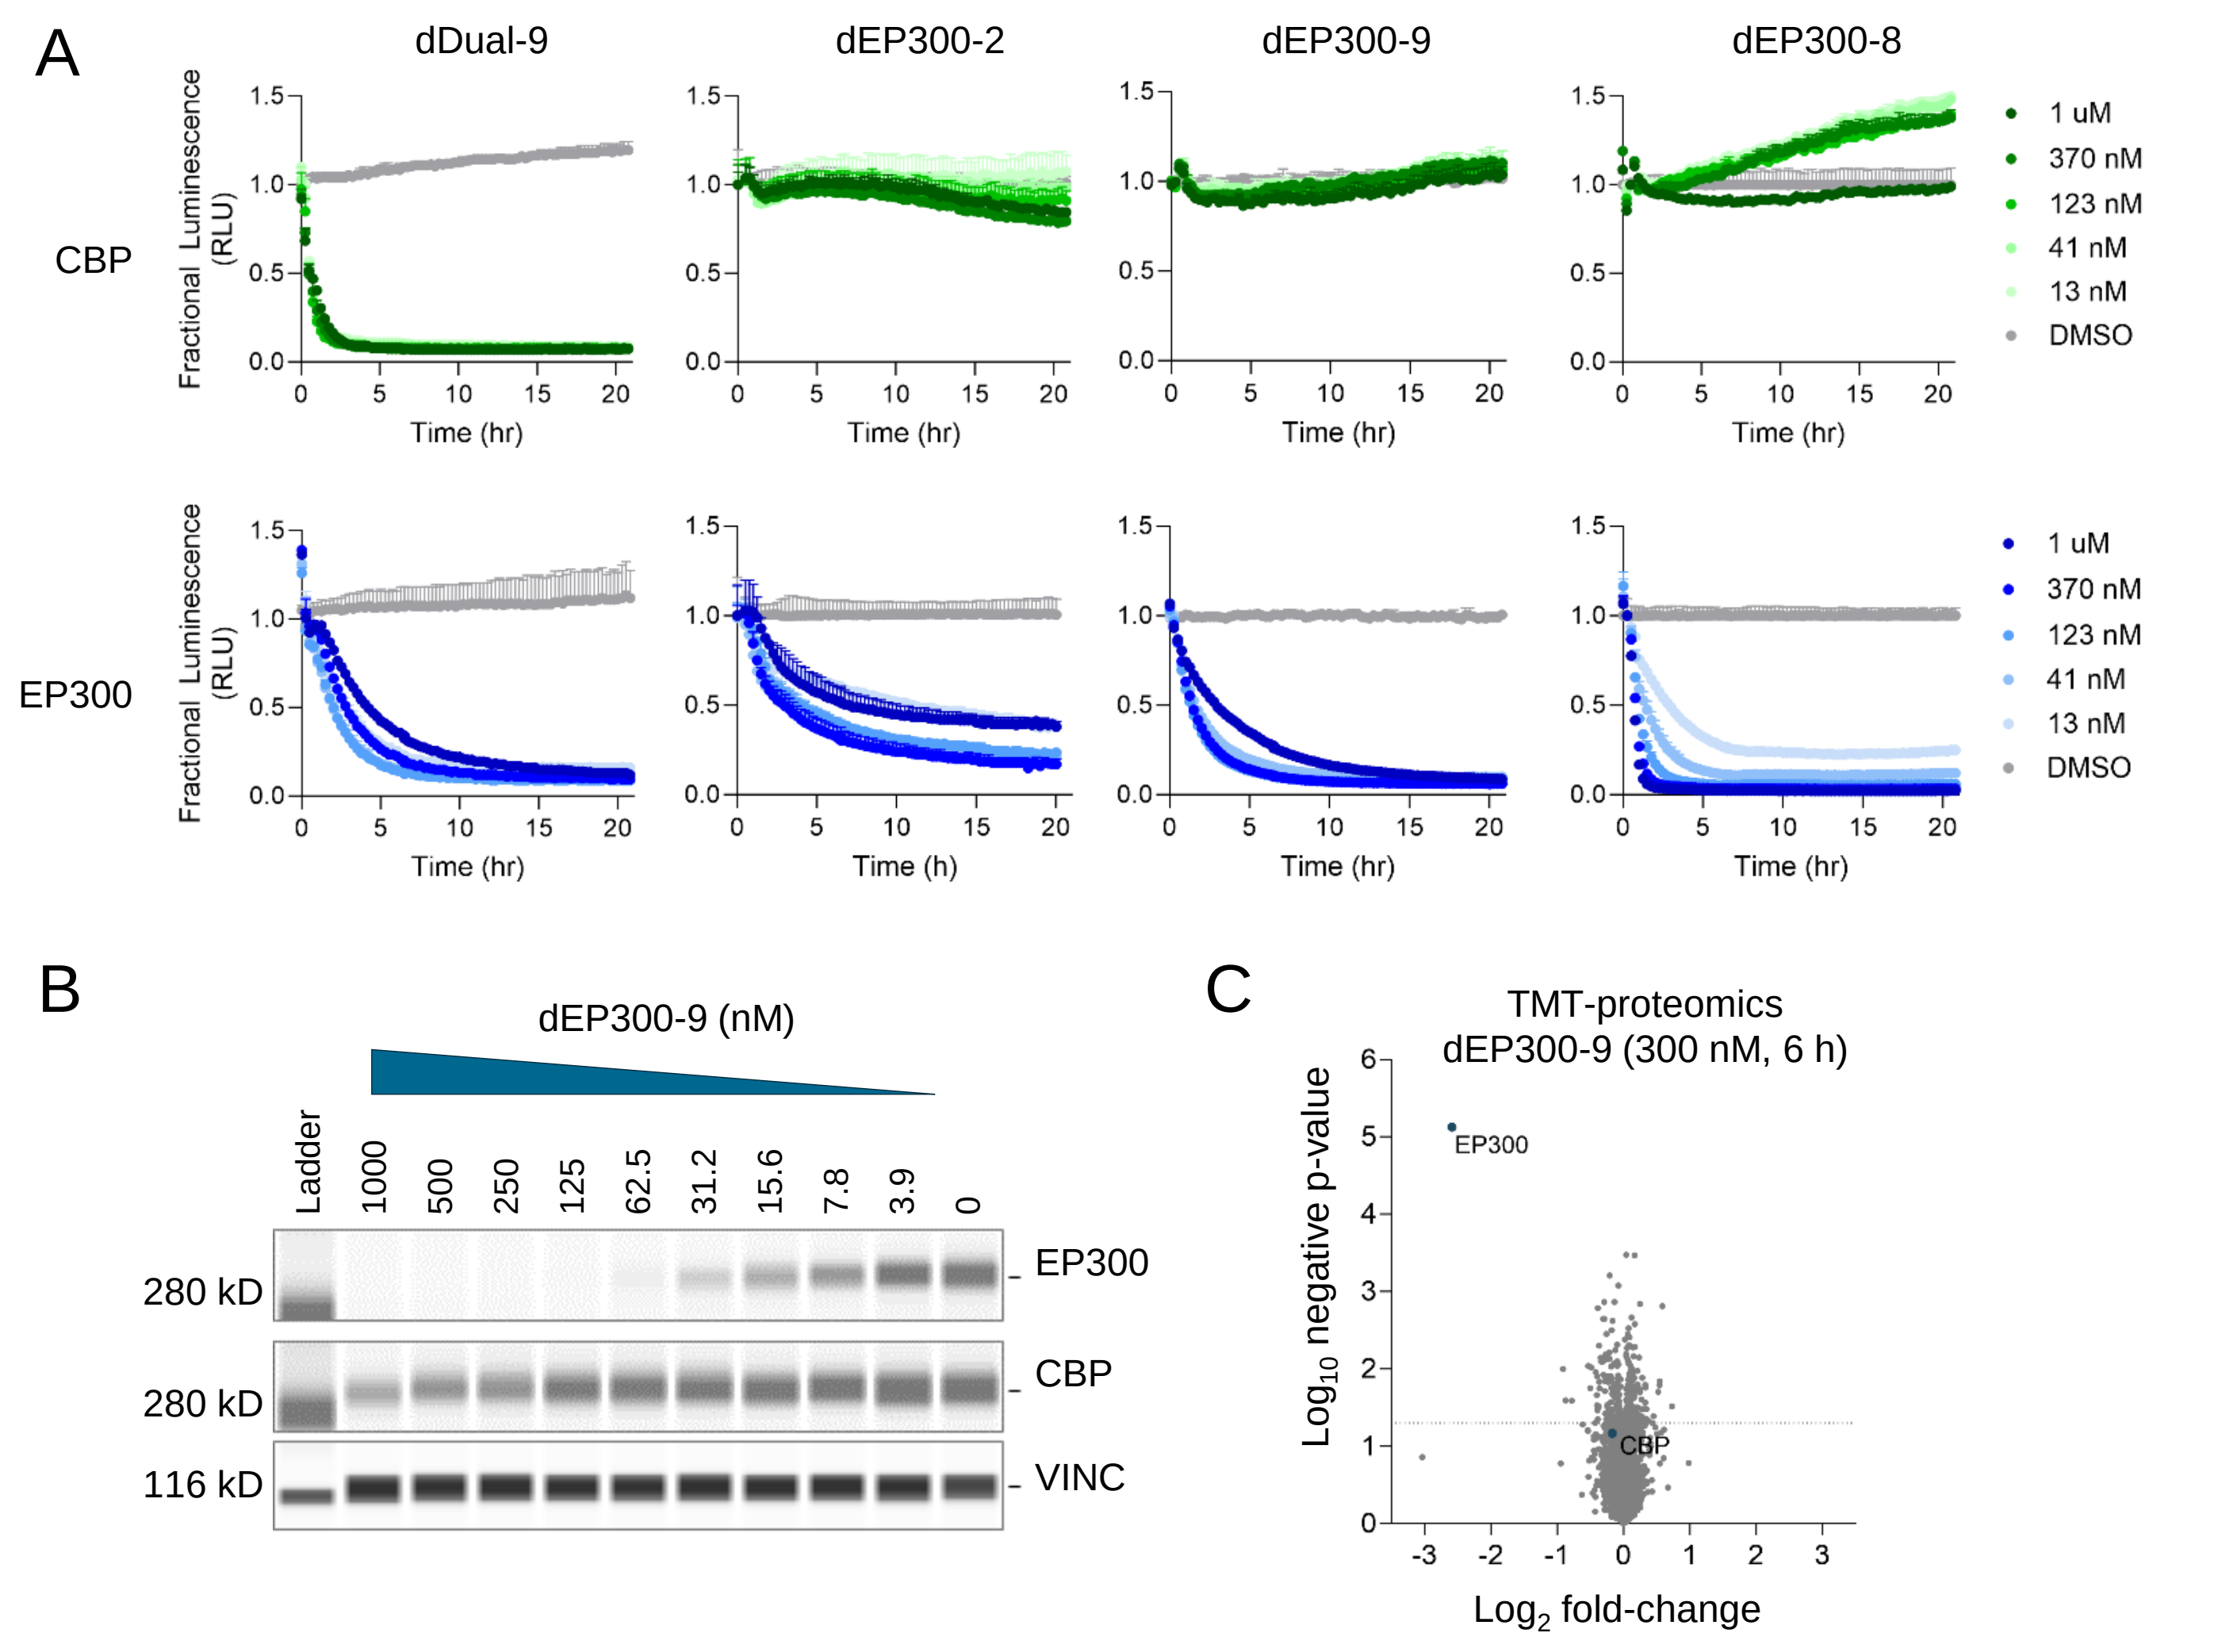
A
dDual-9
dEP300-2
dEP300-9
dEP300-8
CBP
EP300
Fractional Luminescence (RLU)
Time (hr)
1 uM
370 nM
123 nM
41 nM
13 nM
DMSO
B
dEP300-9 (nM)
Ladder
1000
500
250
125
62.5
31.2
15.6
7.8
3.9
0
280 kD
116 kD
EP300
CBP
VINC
C
TMT-proteomics
dEP300-9 (300 nM, 6 h)
Log_{10} negative p-value
Log_2 fold-change

Figure 2: A) Kinetic rates of degradation for dual and EP300-selective degraders in U2OS CBP- and EP300-HiBit knock-in cells (24 h). B) ProteinSimple dose-response western blot for EP300, CBP and Vinculin following 24 h dEP300-9 treatment in Hela cells. C) TMT-proteomics in U2OS cells following dEP300-9 treatment (300 nM) for 6 h.

Figure 3. Degradation of EP300 attenuates androgen signaling

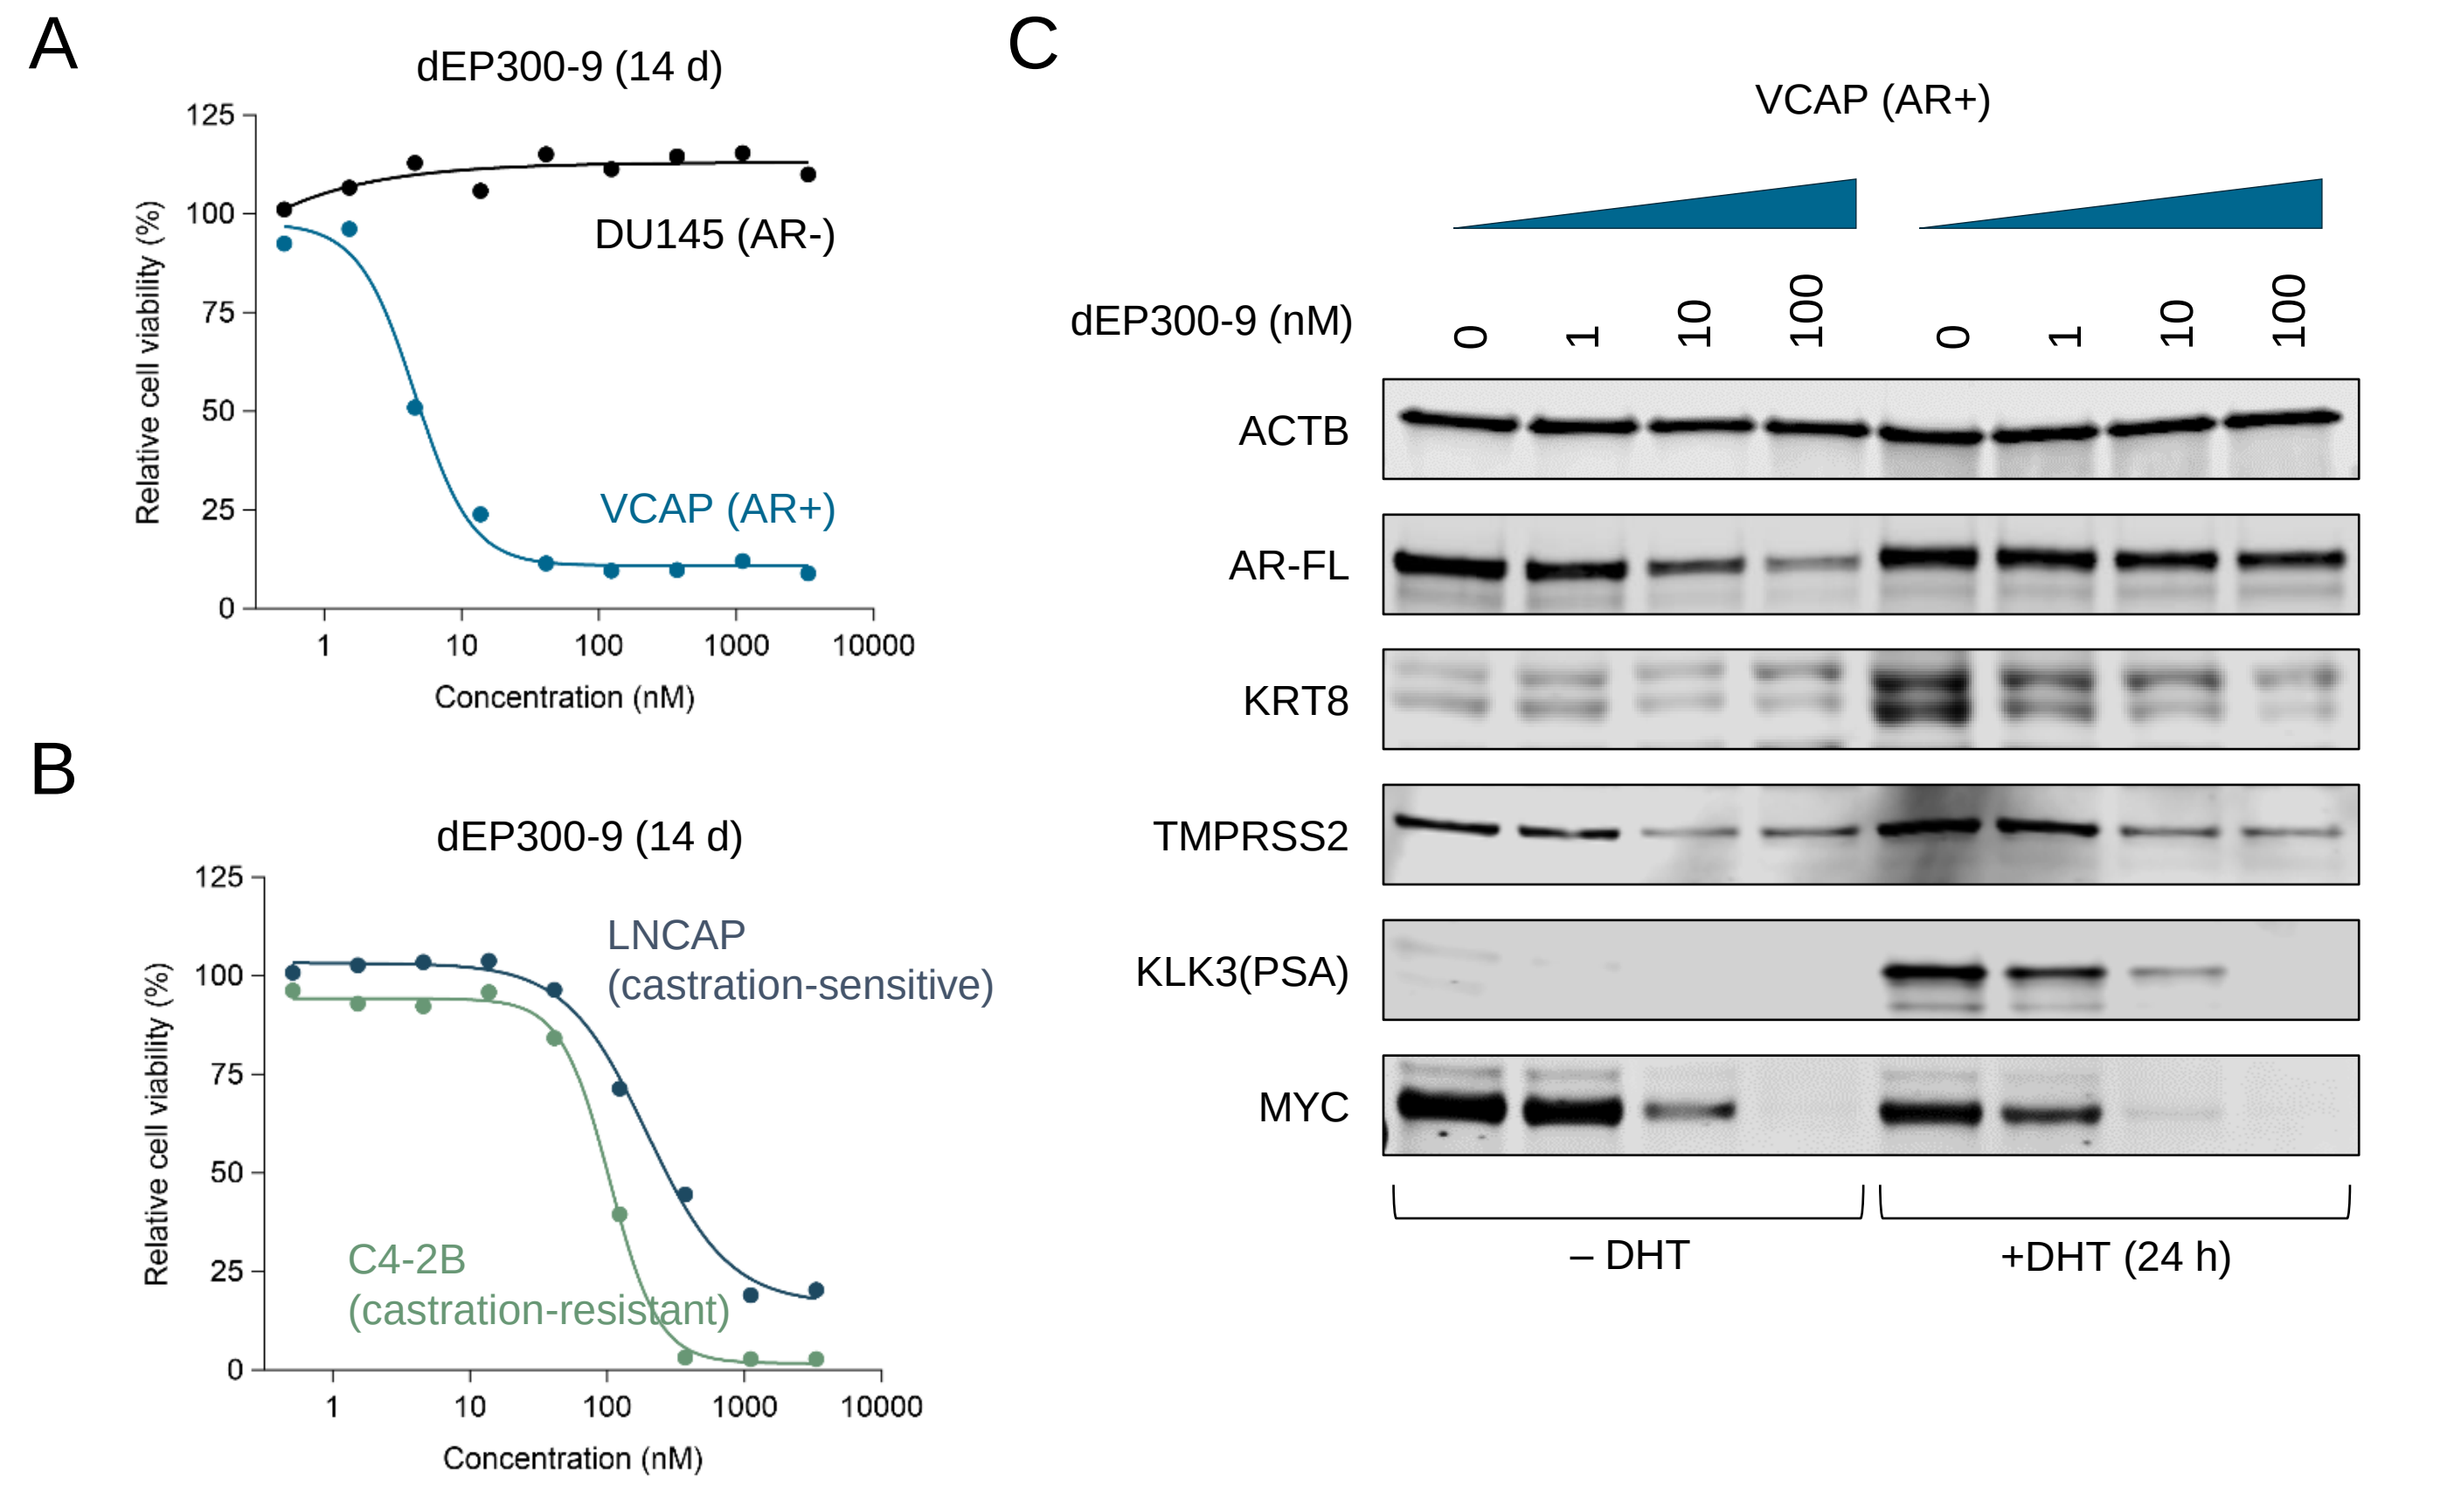
A
dEP300-9 (14 d)
DU145 (AR-)
VCAP (AR+)
Relative cell viability (%)
Concentration (nM)
B
dEP300-9 (14 d)
LNCAP (castration-sensitive)
C4-2B (castration-resistant)
C
VCAP (AR+)
dEP300-9 (nM)
0
1
10
100
ACTB
AR-FL
KRT8
TMPRSS2
KLK3(PSA)
MYC
– DHT
+DHT (24 h)

Figure 3: A) Cell titer glo (cell viability) assay demonstrating that dEP300-9 reduced the growth of VCAP (AR+), but not DU145 (AR-) cells, in a 14 d cell growth assay. B) Cell titer glo (cell viability) assay demonstrating that dEP300-9 reduced the growth of both LNCAP (castration-sensitive) and the clonal cell line C4-2B (castration-resistant) cells, in a 14 d cell growth assay. C) Dose-response western blot for AR-regulated gene products in VCAP cells pre-treated with dEP300-9 for 24 h and stimulated with 10 nM dihydrotestosterone (DHT) for 24 h.

Figure 4. dEP300-2 treatment reduced VCAP xenograft tumor growth

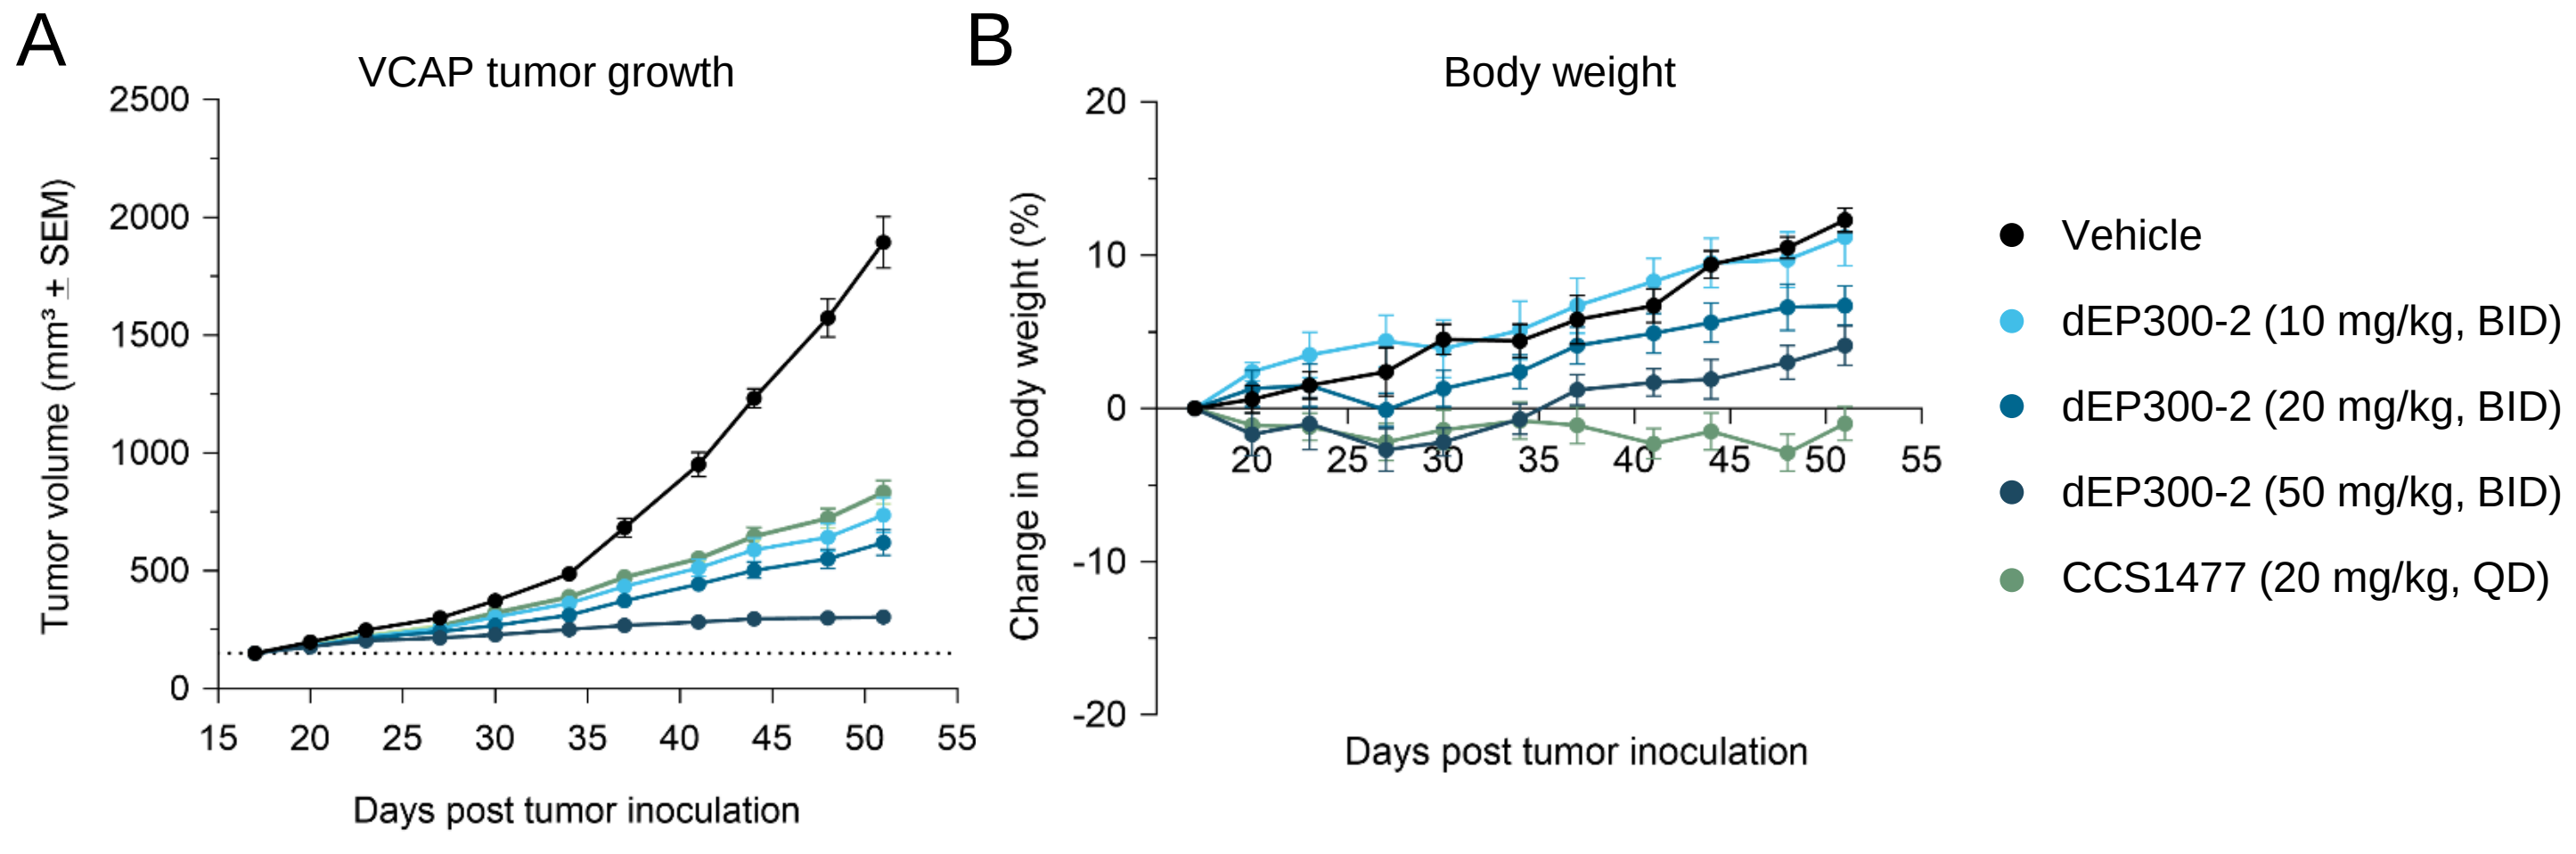
A
VCAP tumor growth
Tumor volume (mm³ ± SEM)
Days post tumor inoculation
B
Body weight
Change in body weight (%)
Days post tumor inoculation
Vehicle
dEP300-2 (10 mg/kg, BID)
dEP300-2 (20 mg/kg, BID)
dEP300-2 (50 mg/kg, BID)
CCS1477 (20 mg/kg, QD)

Figure 4: A) Volume of VCAP tumor xenografts through 52 d post-implantation. Mice were treated with vehicle control (BID), dEP300-2 (10, 20 or 50 mg/kg, BID), or CCS1477 (20 mg/kg, QD) B) Body weight change in tumor-bearing mice treated with vehicle, dEP300-2, or CCS1477 through 52 d post-implantation.

Figure 5. Reduced thrombocytopenia with EP300 degradation compared to dual inhibition

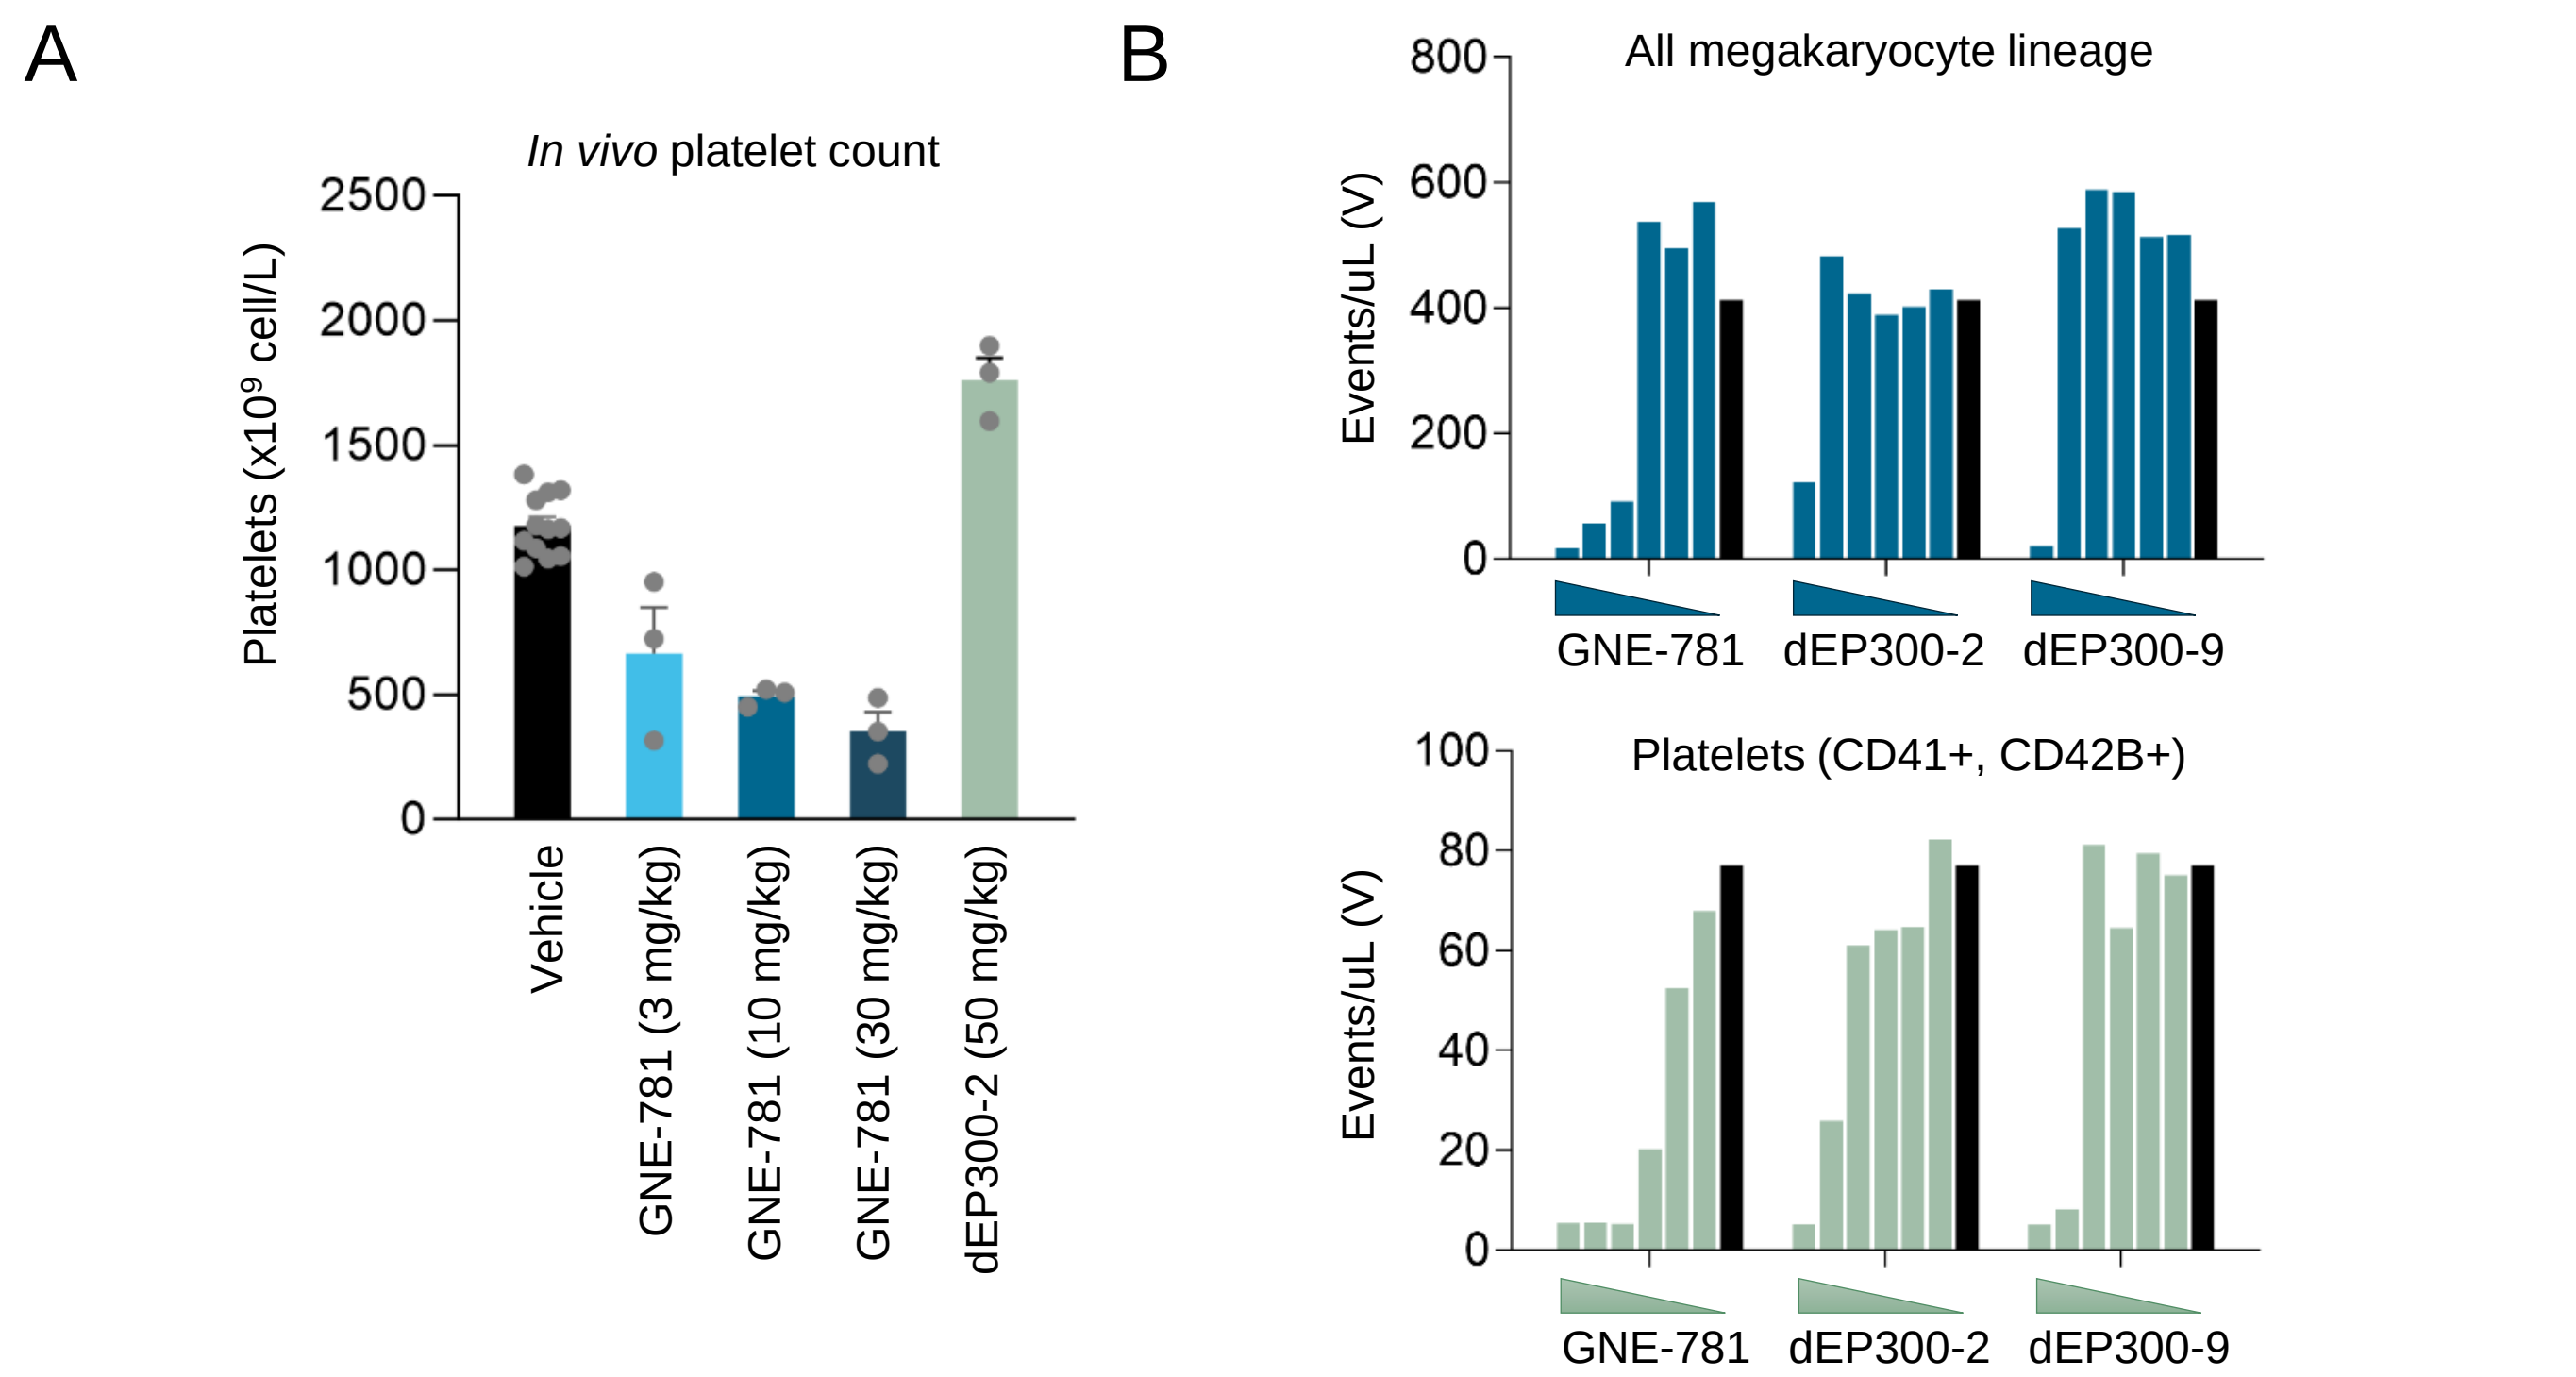
A
In vivo platelet count
Platelets ($x10^9$ cell/L)
Vehicle
GNE-781 (3 mg/kg)
GNE-781 (10 mg/kg)
GNE-781 (30 mg/kg)
dEP300-2 (50 mg/kg)
B
All megakaryocyte lineage
Platelets (CD41+, CD42B+)
Events/uL (V)
GNE-781
dEP300-2
dEP300-9

Figure 5: A) Platelet levels in mice measured after 2 weeks of BID dosing with GNE-781 (3, 10 or 30 mg/kg) or dEP300-2 (50 mg/kg). B) *In vitro* human megakaryocyte differentiation and platelet formation assay demonstrates that dEP300-2 and dEP300-9 treatment results in less cytopenia and thrombocytopenia than dual inhibition with GNE-781 (dose range: 5000 nM – 5 nM).

CONCLUSIONS

- Selective disruption of EP300 while sparing CBP is achievable with targeted protein degradation techniques
- EP300 is an essential mediator of androgen signaling in prostate adenocarcinoma and is required for cell growth and survival in this disease
- Unlike dual inhibition, EP300 degradation does not have a strong effect on platelet levels and megakaryocyte viability in mice and *ex vivo* human PBMCs
- Targeting EP300 through selective protein degradation may provide a safer and more effective treatment strategy for advanced cancers

ACKNOWLEDGEMENTS: The team owes many thanks to Ammar Adam, GiNell Elliot, Janice Lee, Solymar Negretti, Matt Netherton, Brenna Sherbanee, Lee Silverman and David Terry for their support and contributions.