IDENTIFICATION OF SELECTIVE CBP DEGRADERS WITH ROBUST PRECLINICAL PK/PD, EFFICACY AND SAFETY ACROSS SOLID TUMOR INDICATIONS

Darshan Sappal, Benjamin Adams, Ketaki Adhikari, Wesley Austin, Breanna Bullock, Danette Daniels, Julie Di Bernardo, Thomas Dixon, Brian Ethell, Md Imran Hossain, Laura La Bonte, Dave Lahr, Mei Yun Lin, Karolina Mizeracka, Paige Monsen, Shawn Schiller, Nihan Ucisik, Grace Werosta, Molly Wilson, Elizabeth Wittenborn, Mark Zimmerman

FOGHORN® THERAPEUTICS

ABSTRACT

CREB binding protein (CBP) and E1A binding protein p300 (EP300) are paralog histone acetyltransferases involved in many cellular processes via their activity as transcription factor co-activators. Dysregulation of one or both proteins has been implicated in various cancer types, and functional genomic screens have revealed a bidirectional synthetic lethal relationship between these two paralogs in tumor cells. Due to the high homology between CBP and EP300, identifying selective chemical matter that selectively disrupts the activity of CBP has proven challenging. Small molecule inhibitors targeting the HAT or bromodomain of CBP/EP300 have been developed; however, these agents exhibit hematopoietic toxicity resulting from dual inhibition, which limits their therapeutic window.

Herein, we describe the PK, PD, and efficacy of selective, potent CBP degraders across various EP300-mutant cancer xenograft models. Our results show deep and sustained CBP degradation, leading to significant tumor growth inhibition in solid tumors. This anti-tumor activity was not associated with significant body weight loss or hematopoietic toxicity. Our CBP-selective protein degraders have the potential to be a first-in-class therapeutic option for patients with tumors harboring EP300 mutations.

Figure 1. Targeting CBP in EP300 mutant cancers

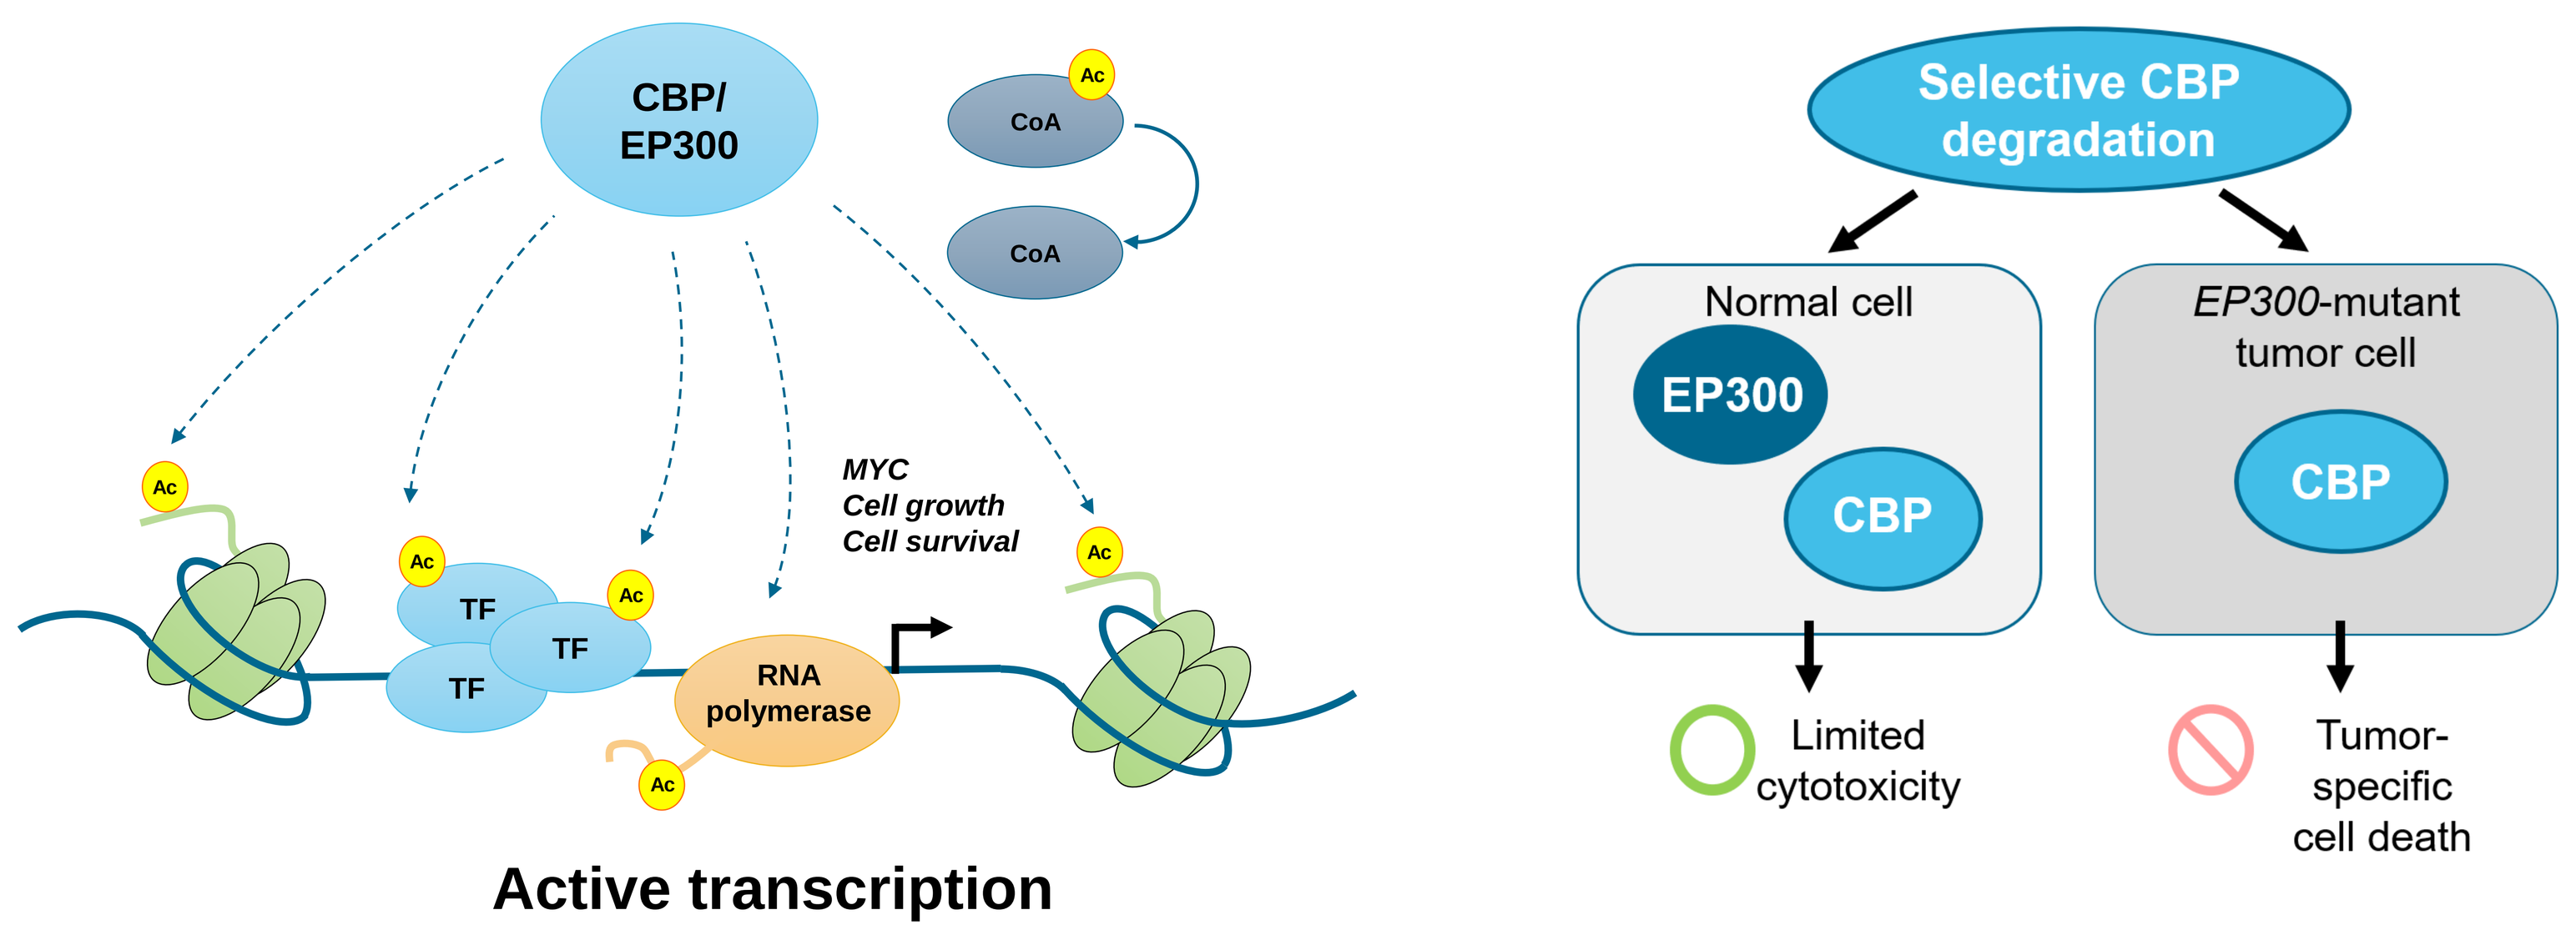
CBP/ EP300
CoA
Ac
MYC
Cell growth
Cell survival
TF
RNA polymerase
Active transcription
Selective CBP degradation
Normal cell
EP300
CBP
EP300-mutant tumor cell
Limited cytotoxicity
Tumor-specific cell death

Figure 1: CBP/EP300 are histone acetyltransferases that function by regulating enhancer mediated transcription and protein stability. These paralogs have a bidirectional synthetic lethal relationship that can be exploited in a variety of cancer indications.

Figure 2. Identification of potent, selective CBP degraders

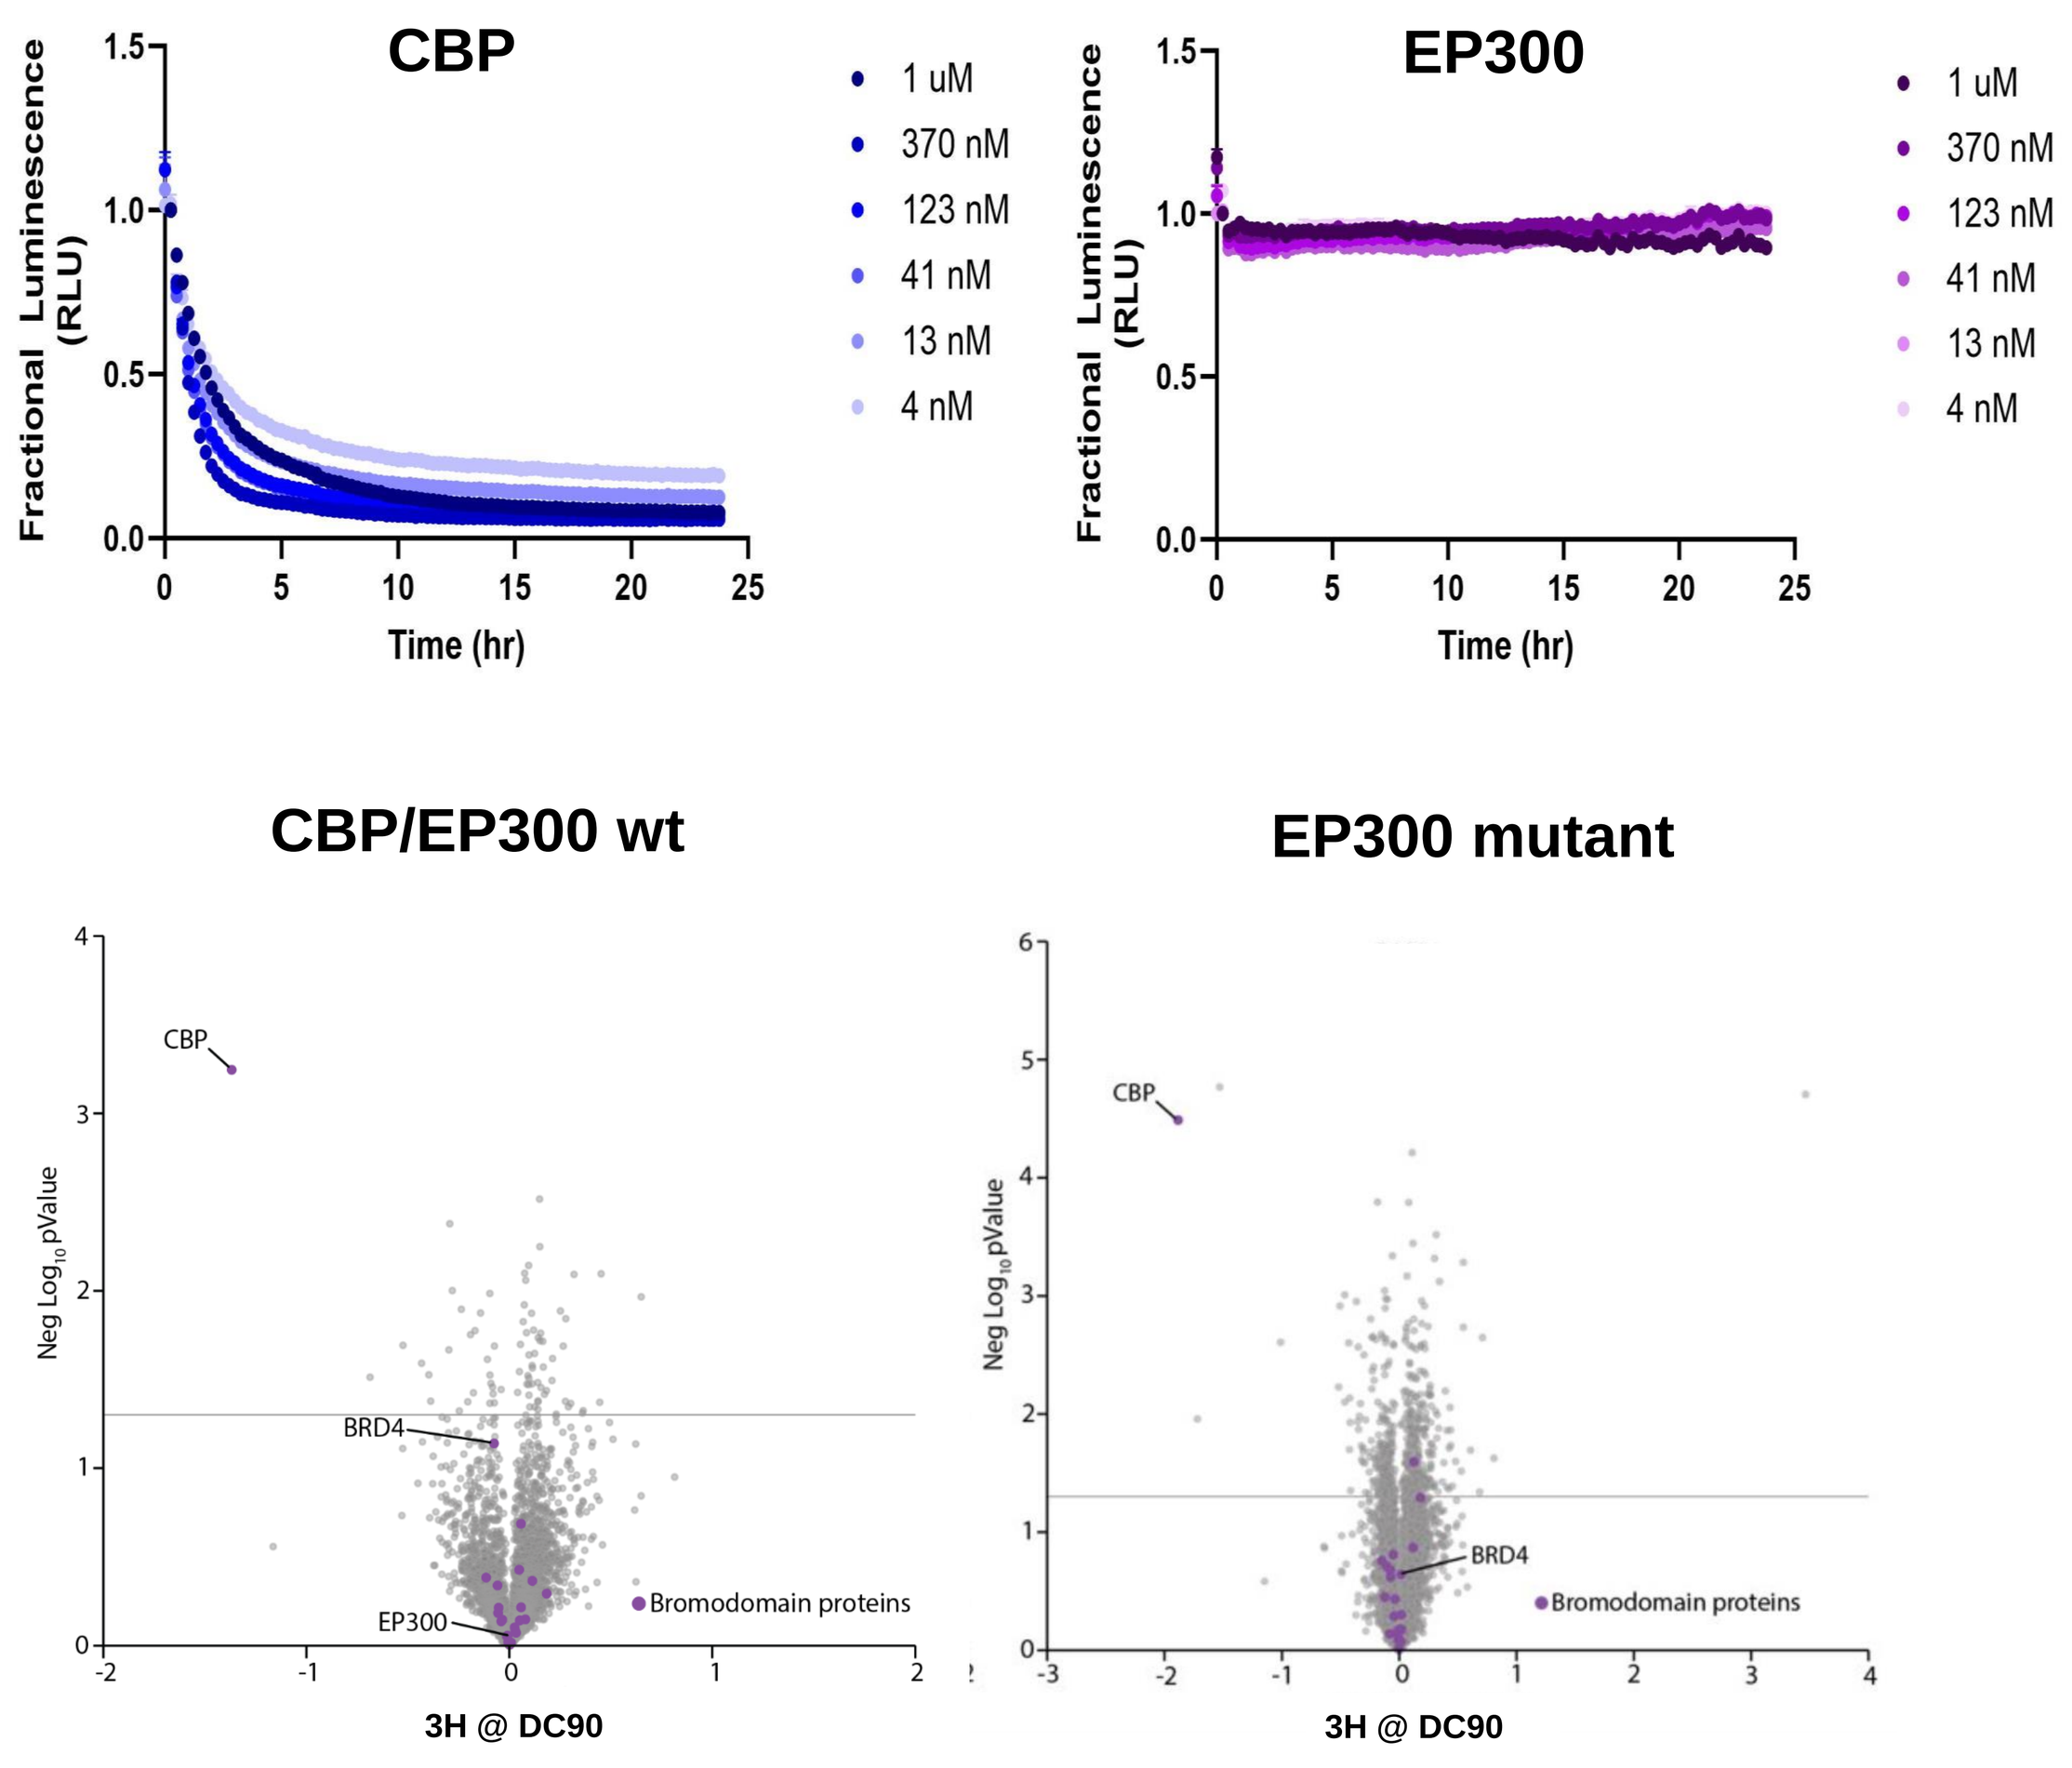
CBP
EP300
1 uM
370 nM
123 nM
41 nM
13 nM
4 nM
Fractional Luminescence (RLU)
Time (hr)
CBP/EP300 wt
EP300 mutant
CBP
BRD4
EP300
Bromodomain proteins
3H @ DC90

In Vitro Properties	dCBP-63
Cellular Potency DC_{50} (KI HiBiT)	>30 µM (EP300) **0.043 µM (CBP)**
Dmax (KI HiBiT)	0% (EP300) **90% (CBP)**
Kinetic Degradation Rate CBP (I) (hr-1)	1.0
Cellular Proliferation GI50 (µM) – RKO (EP300 mutant)	0.02

Figure 2: We have identified potent and selective degraders of CBP. Dose response kinetic data confirm CBP selectivity, with minimal effects on EP300 and other bromodomain containing proteins.

Figure 3. CBP selective degraders show an antiproliferative effect in EP300 mutant versus wild-type cell lines across indications

3A

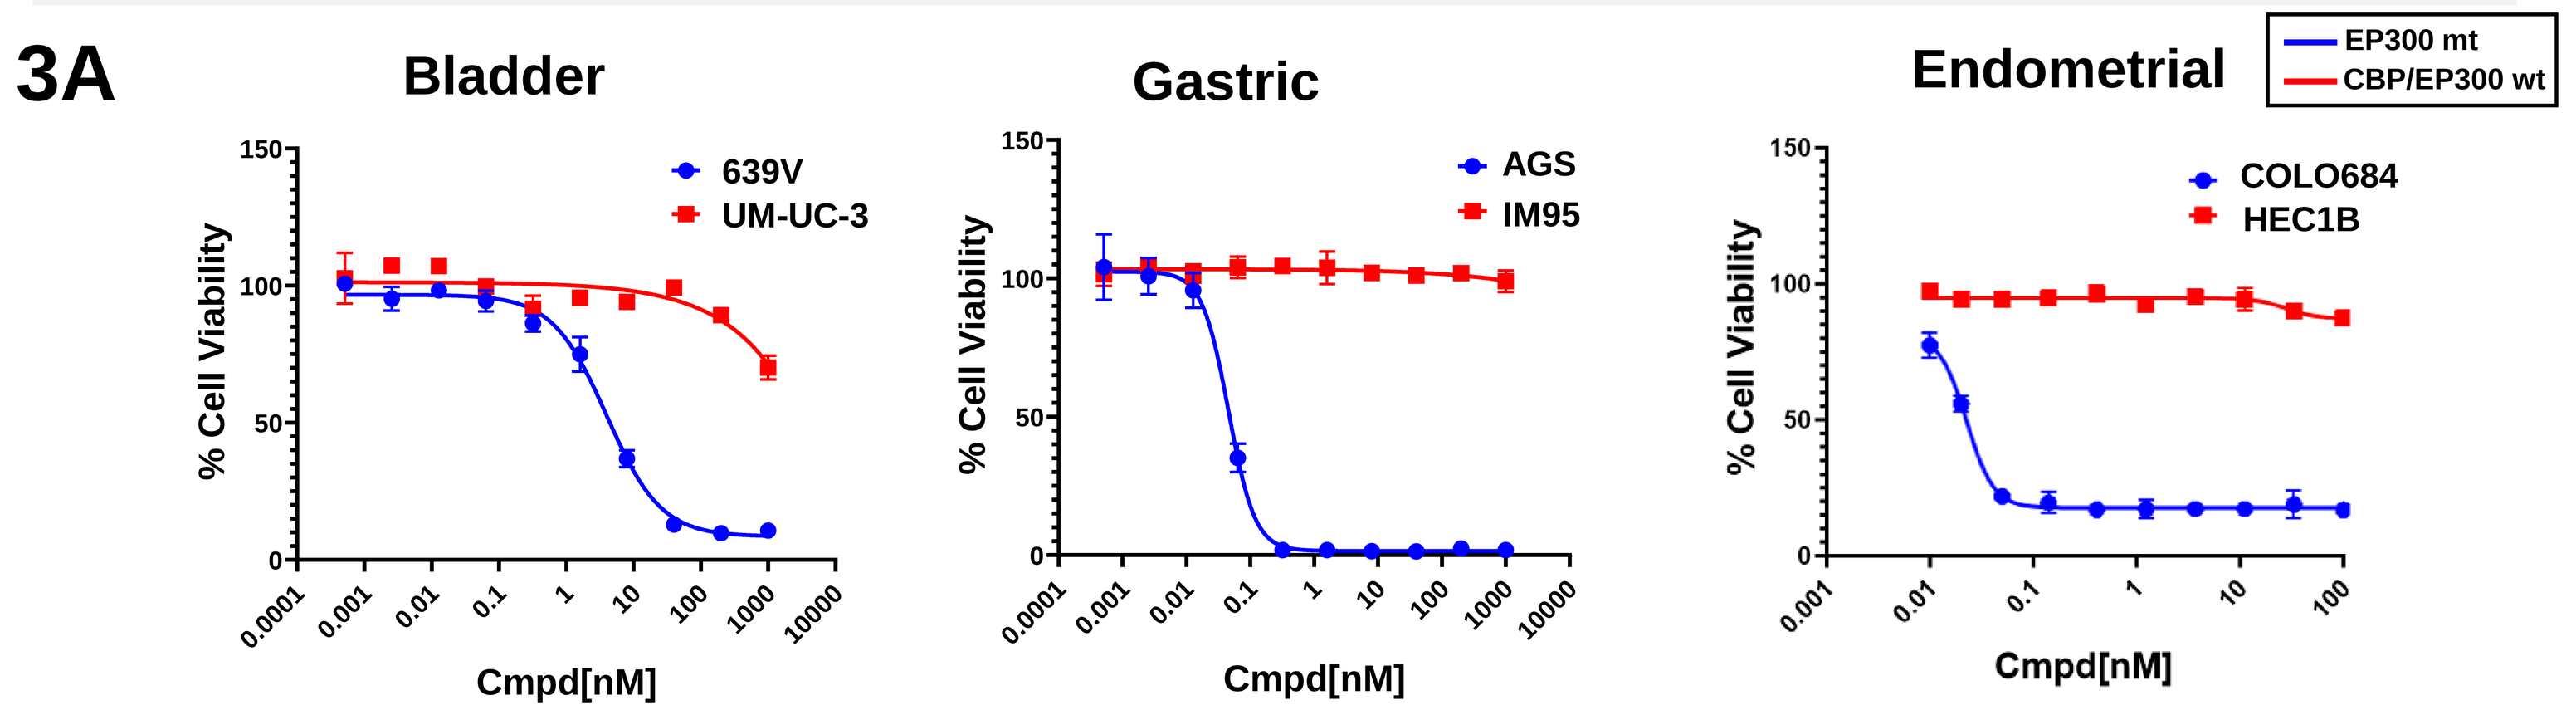
Bladder
Gastric
Endometrial
EP300 mt
CBP/EP300 wt
639V
UM-UC-3
AGS
IM95
COLO684
HEC1B
% Cell Viability
Cmpd[nM]

Figure 3A: Validation of the synthetic lethal effect in EP300 mutant cell lines (blue) while sparing the CBP/EP300 wild-type lines (red) across multiple indications in a 7 day CTG assay.

3B

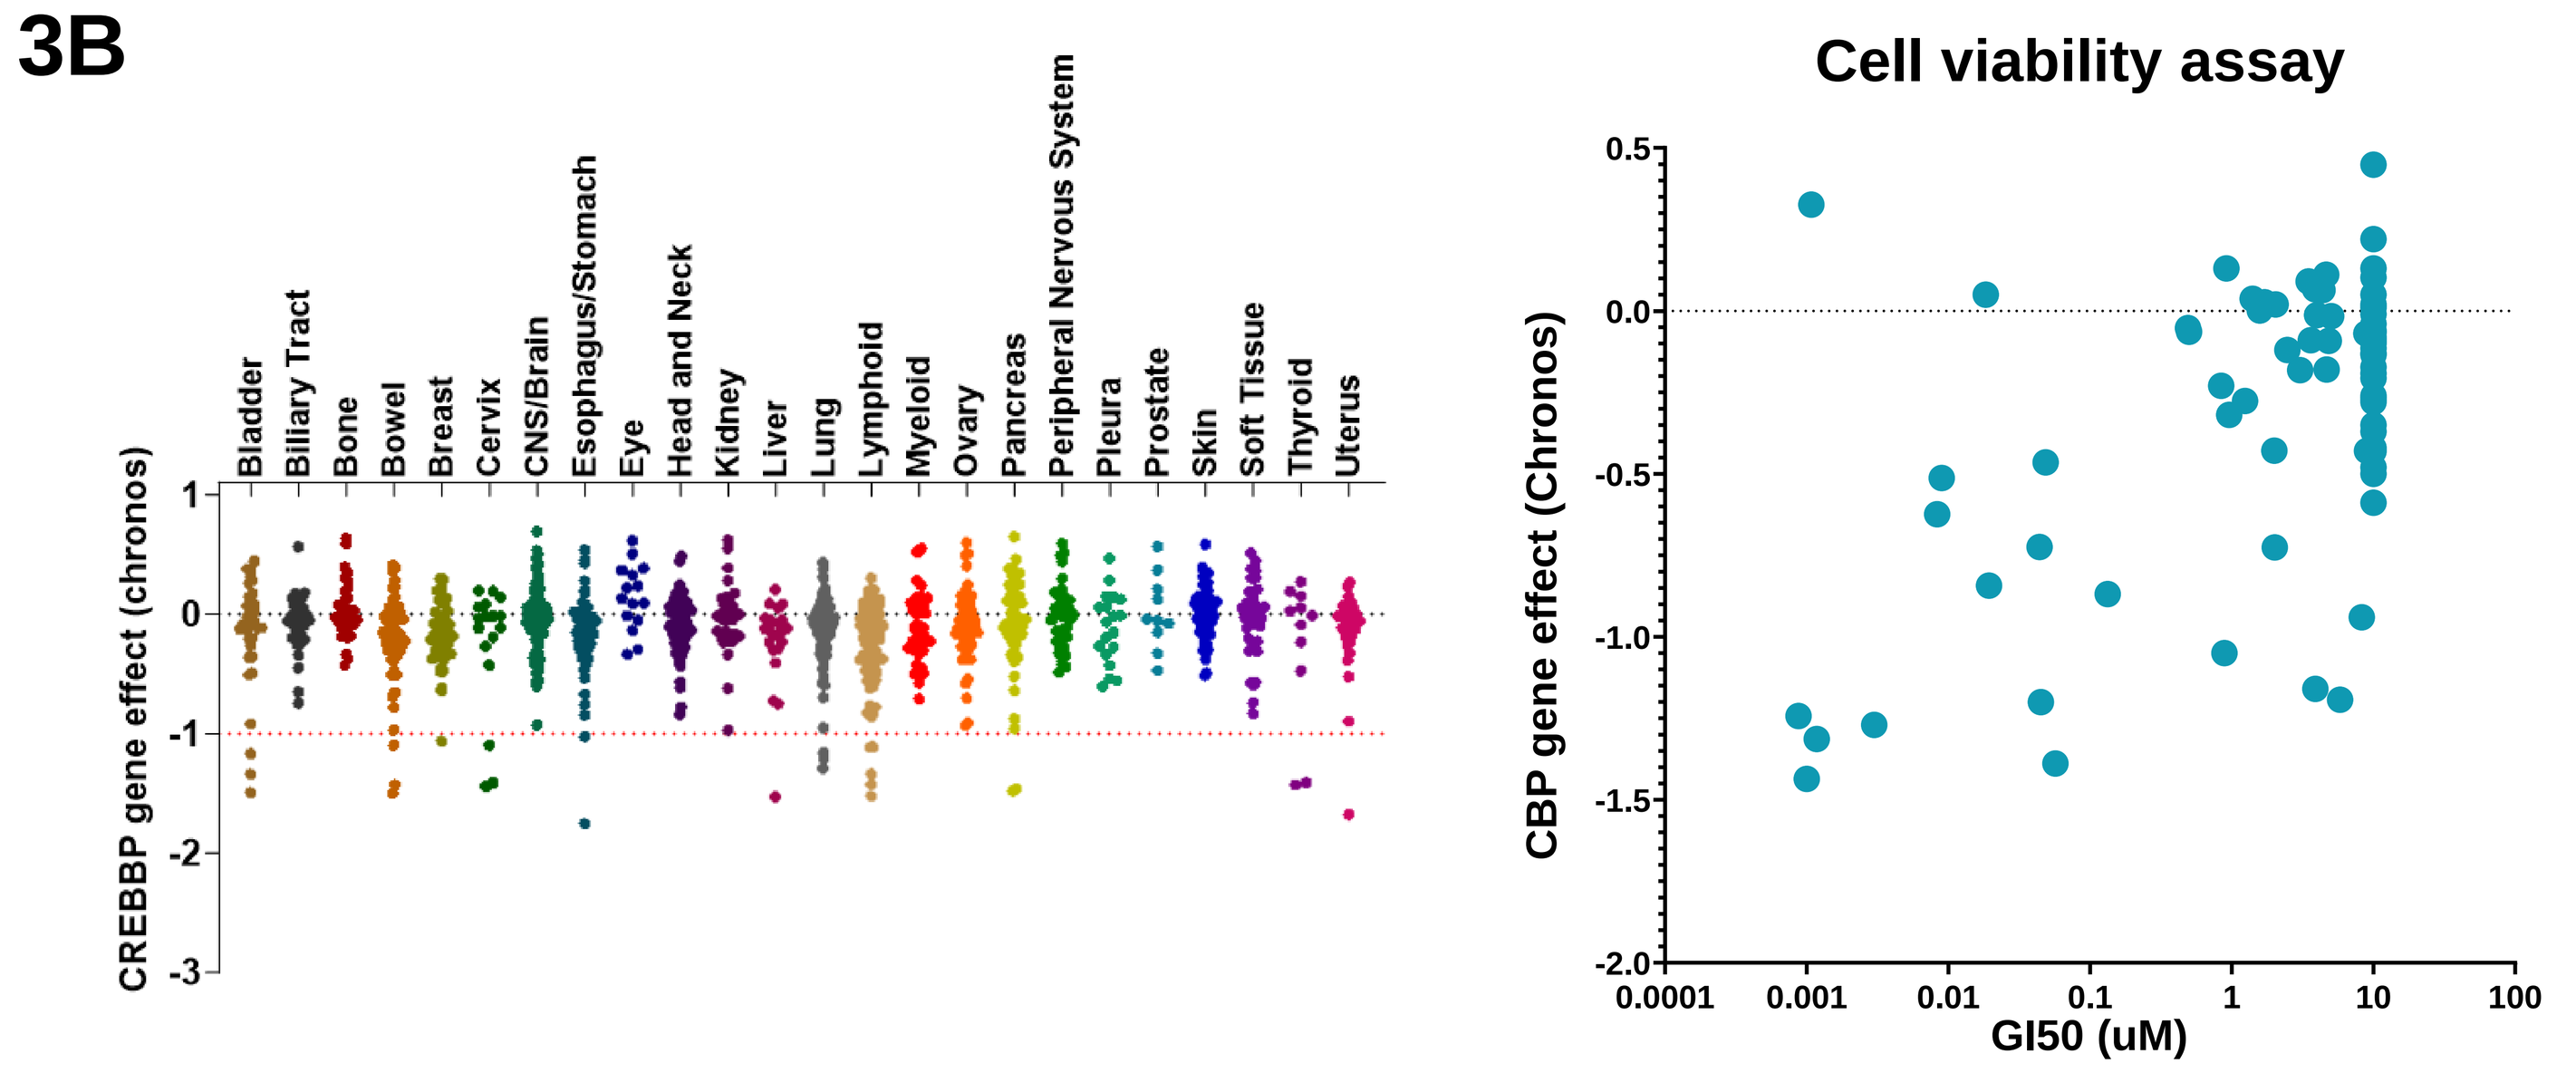
CREBBP gene effect (chronos)
Cell viability assay
CBP gene effect (Chronos)
GI50 (uM)

Figure 3B: Validation of the genetic CBP dependency using our selective degrader across many cell lines from multiple indications of interest.

Figure 4. In vivo PK/PD selectivity and efficacy

4A

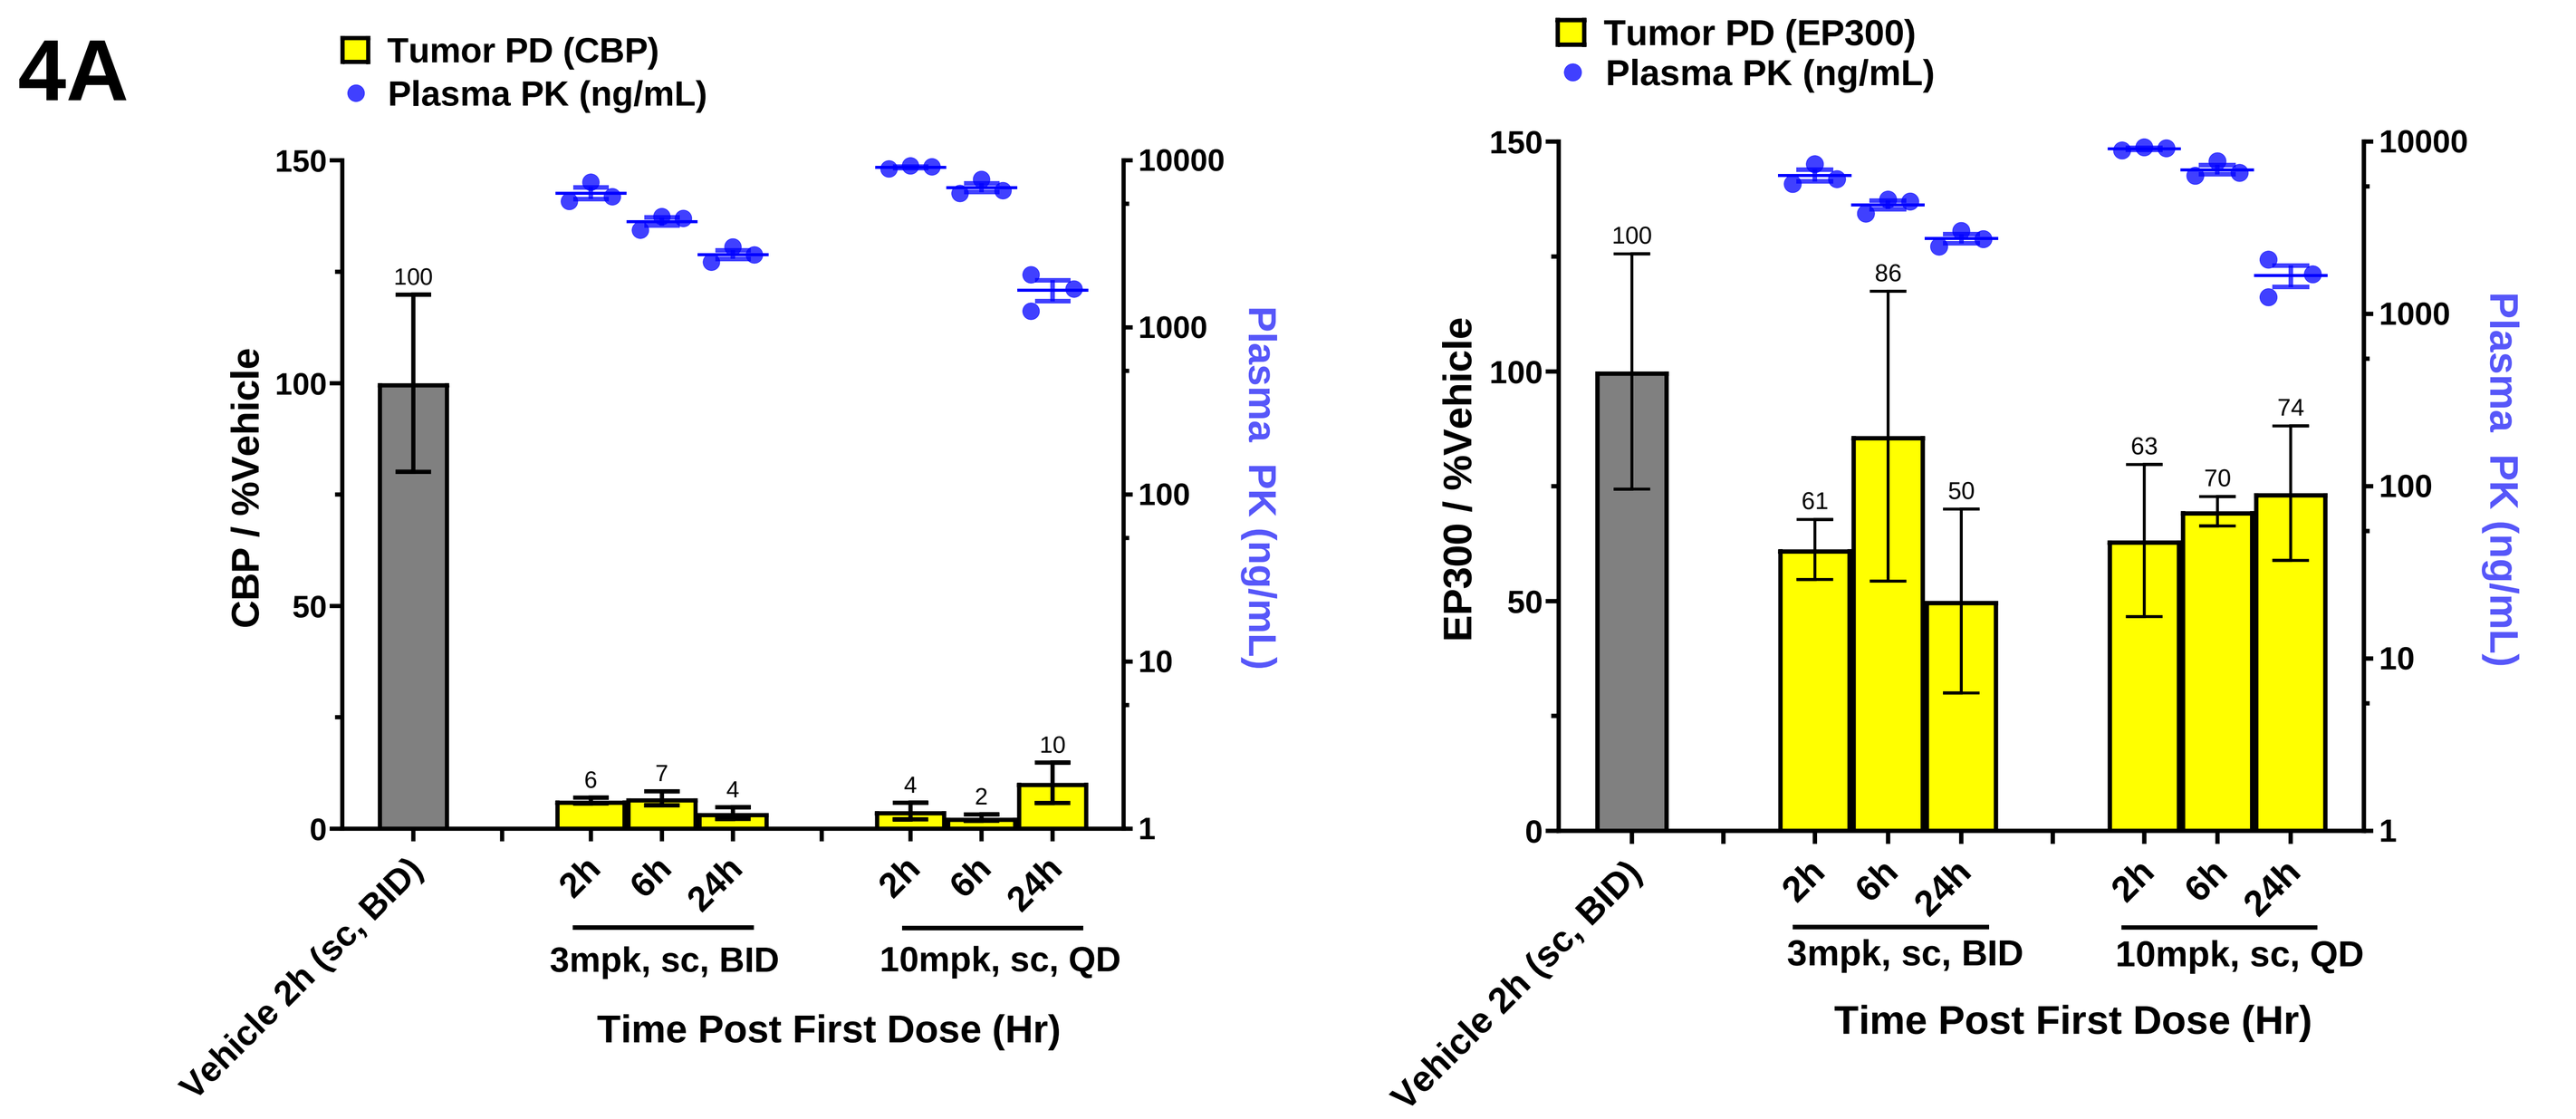
Tumor PD (CBP)
Plasma PK (ng/mL)
Tumor PD (EP300)
CBP / %Vehicle
EP300 / %Vehicle
Vehicle 2h (sc, BID)
3mpk, sc, BID
10mpk, sc, QD
Time Post First Dose (Hr)

Figure 4A: Our in vivo enabled CBP selective degrader, dCBP-59, maintains similar degradation selectivity in vivo as observed in our in vitro assays.

4B

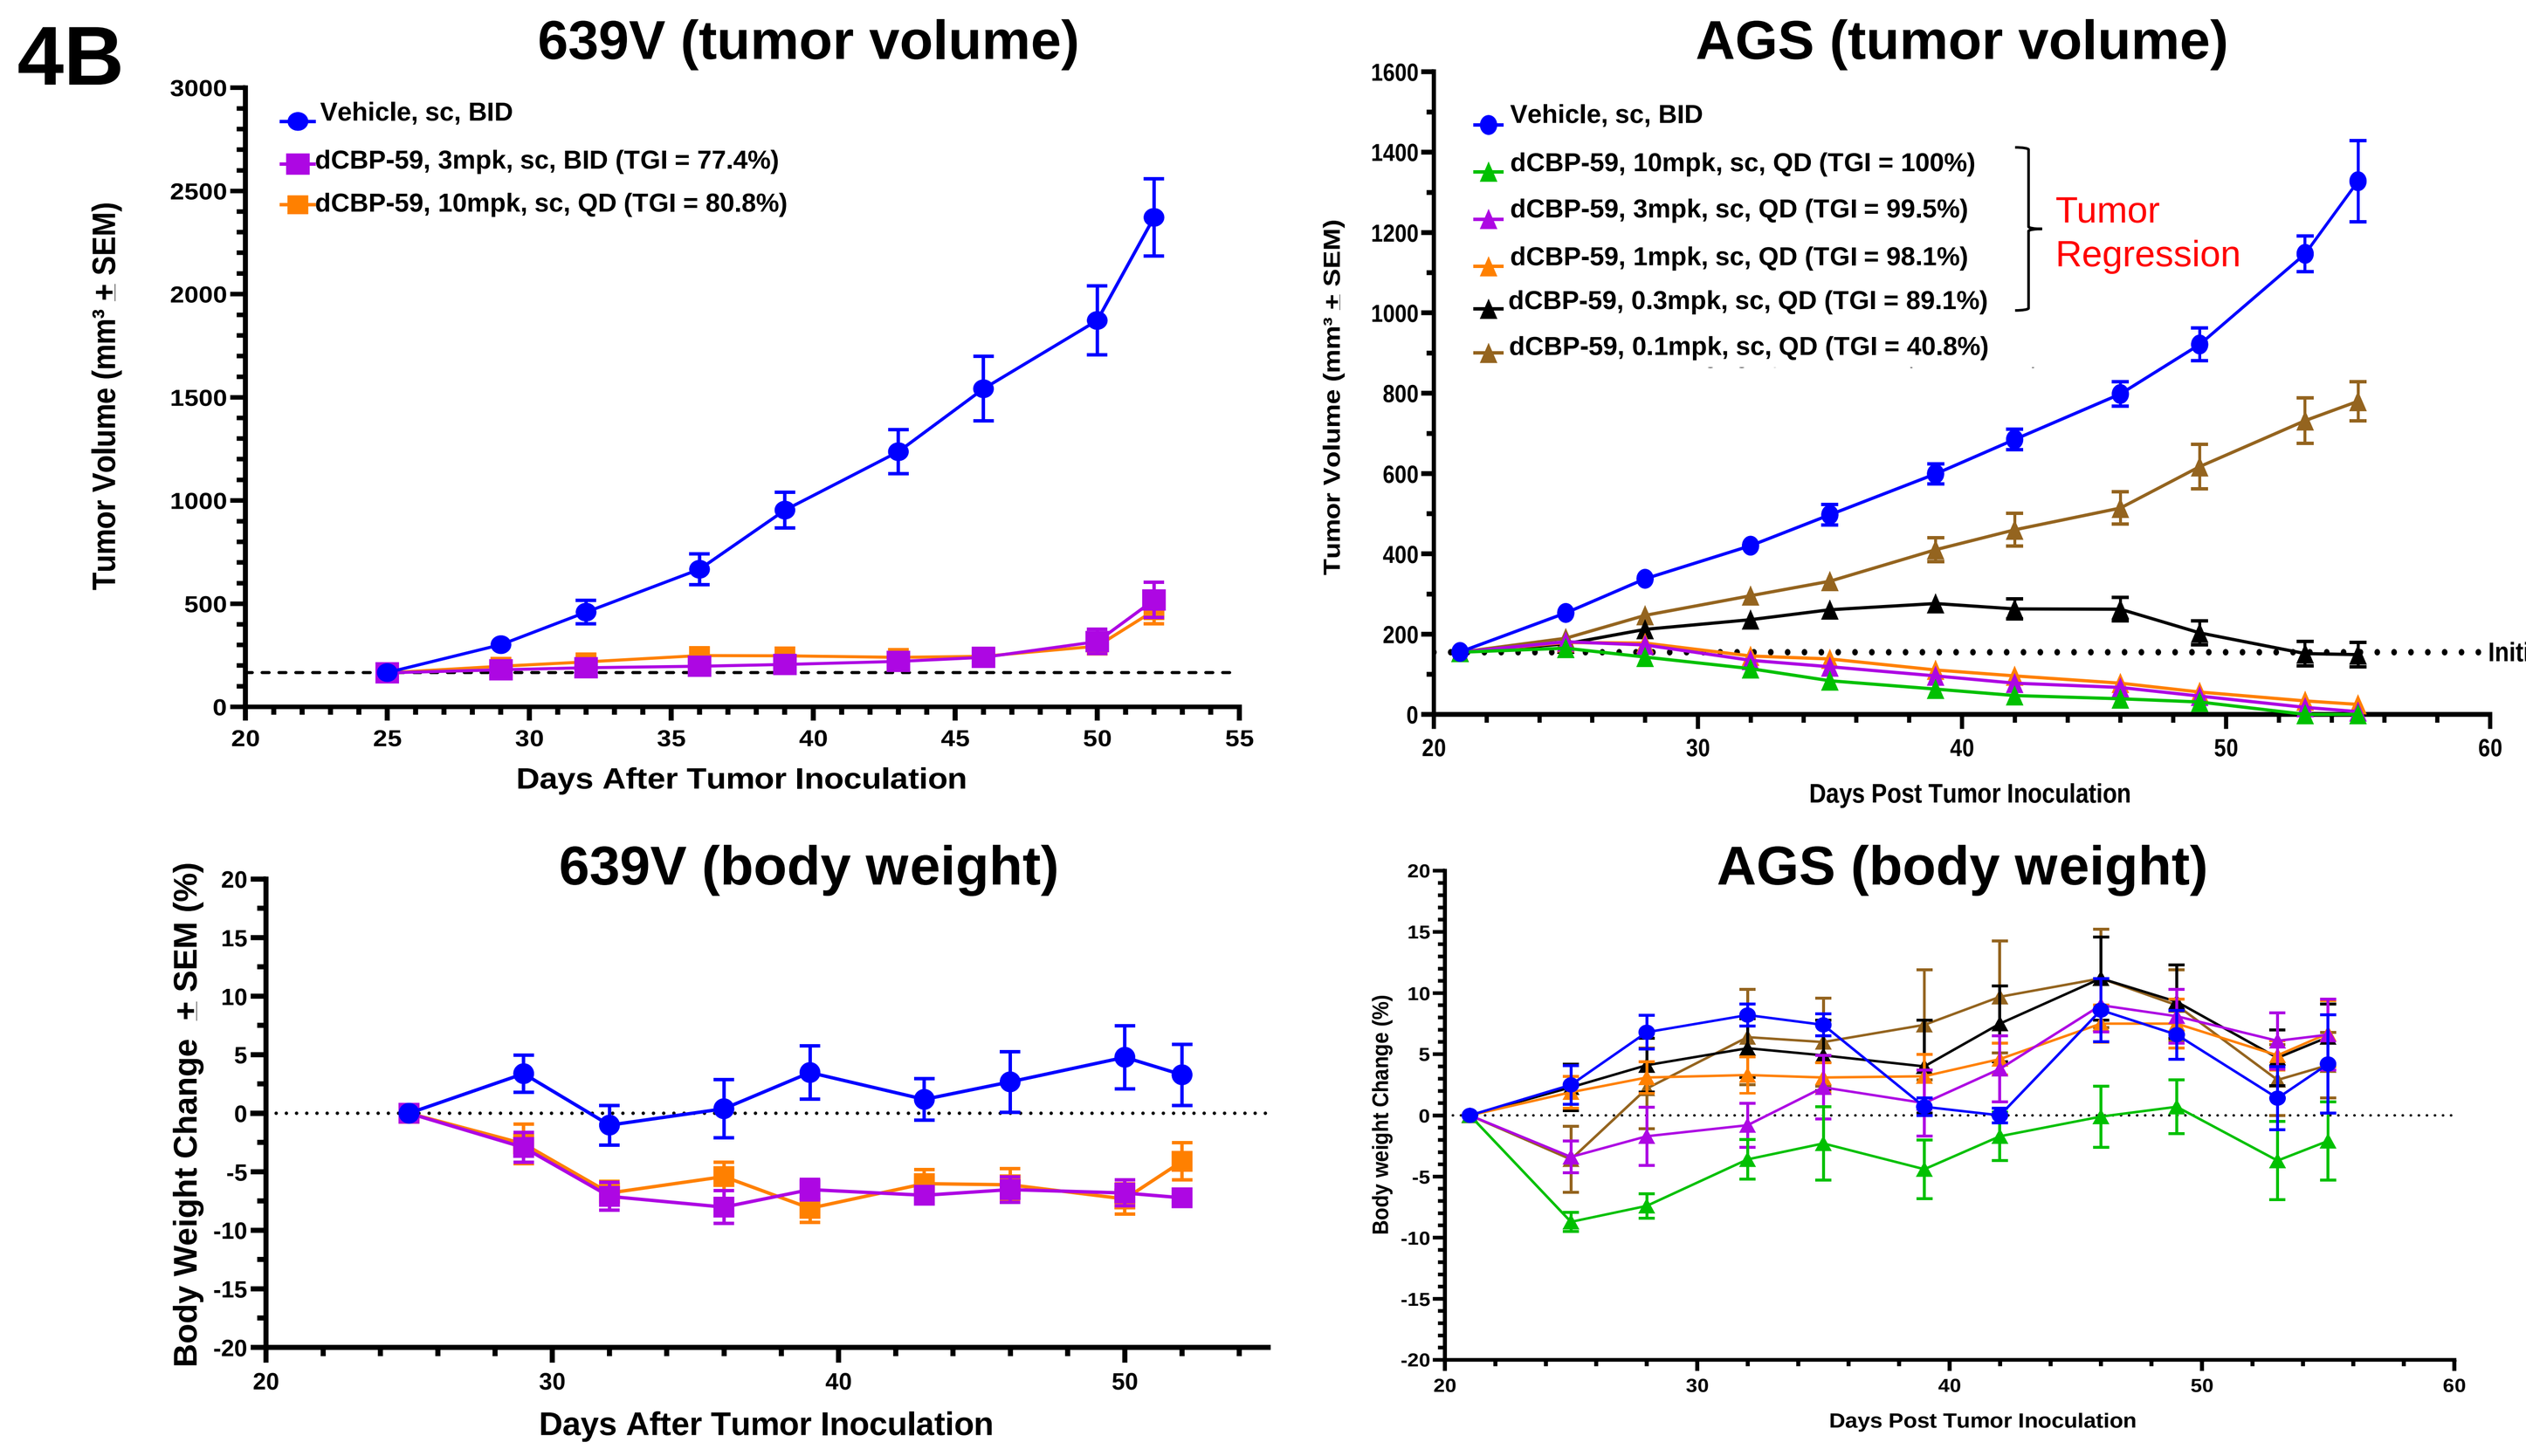
639V (tumor volume)
Vehicle, sc, BID
dCBP-59, 3mpk, sc, BID (TGI = 77.4%)
dCBP-59, 10mpk, sc, QD (TGI = 80.8%)
AGS (tumor volume)
dCBP-59, 10mpk, sc, QD (TGI = 100%)
dCBP-59, 3mpk, sc, QD (TGI = 99.5%)
dCBP-59, 1mpk, sc, QD (TGI = 98.1%)
dCBP-59, 0.3mpk, sc, QD (TGI = 89.1%)
dCBP-59, 0.1mpk, sc, QD (TGI = 40.8%)
Tumor Regression
Initial TV Start
639V (body weight)
AGS (body weight)
Days After Tumor Inoculation
Days Post Tumor Inoculation

Figure 4B: We have achieved excellent efficacy in a number of different EP300 mutant xenograft models across multiple indications.

Figure 5. Degrader impact on H3K27Ac mediated transcription

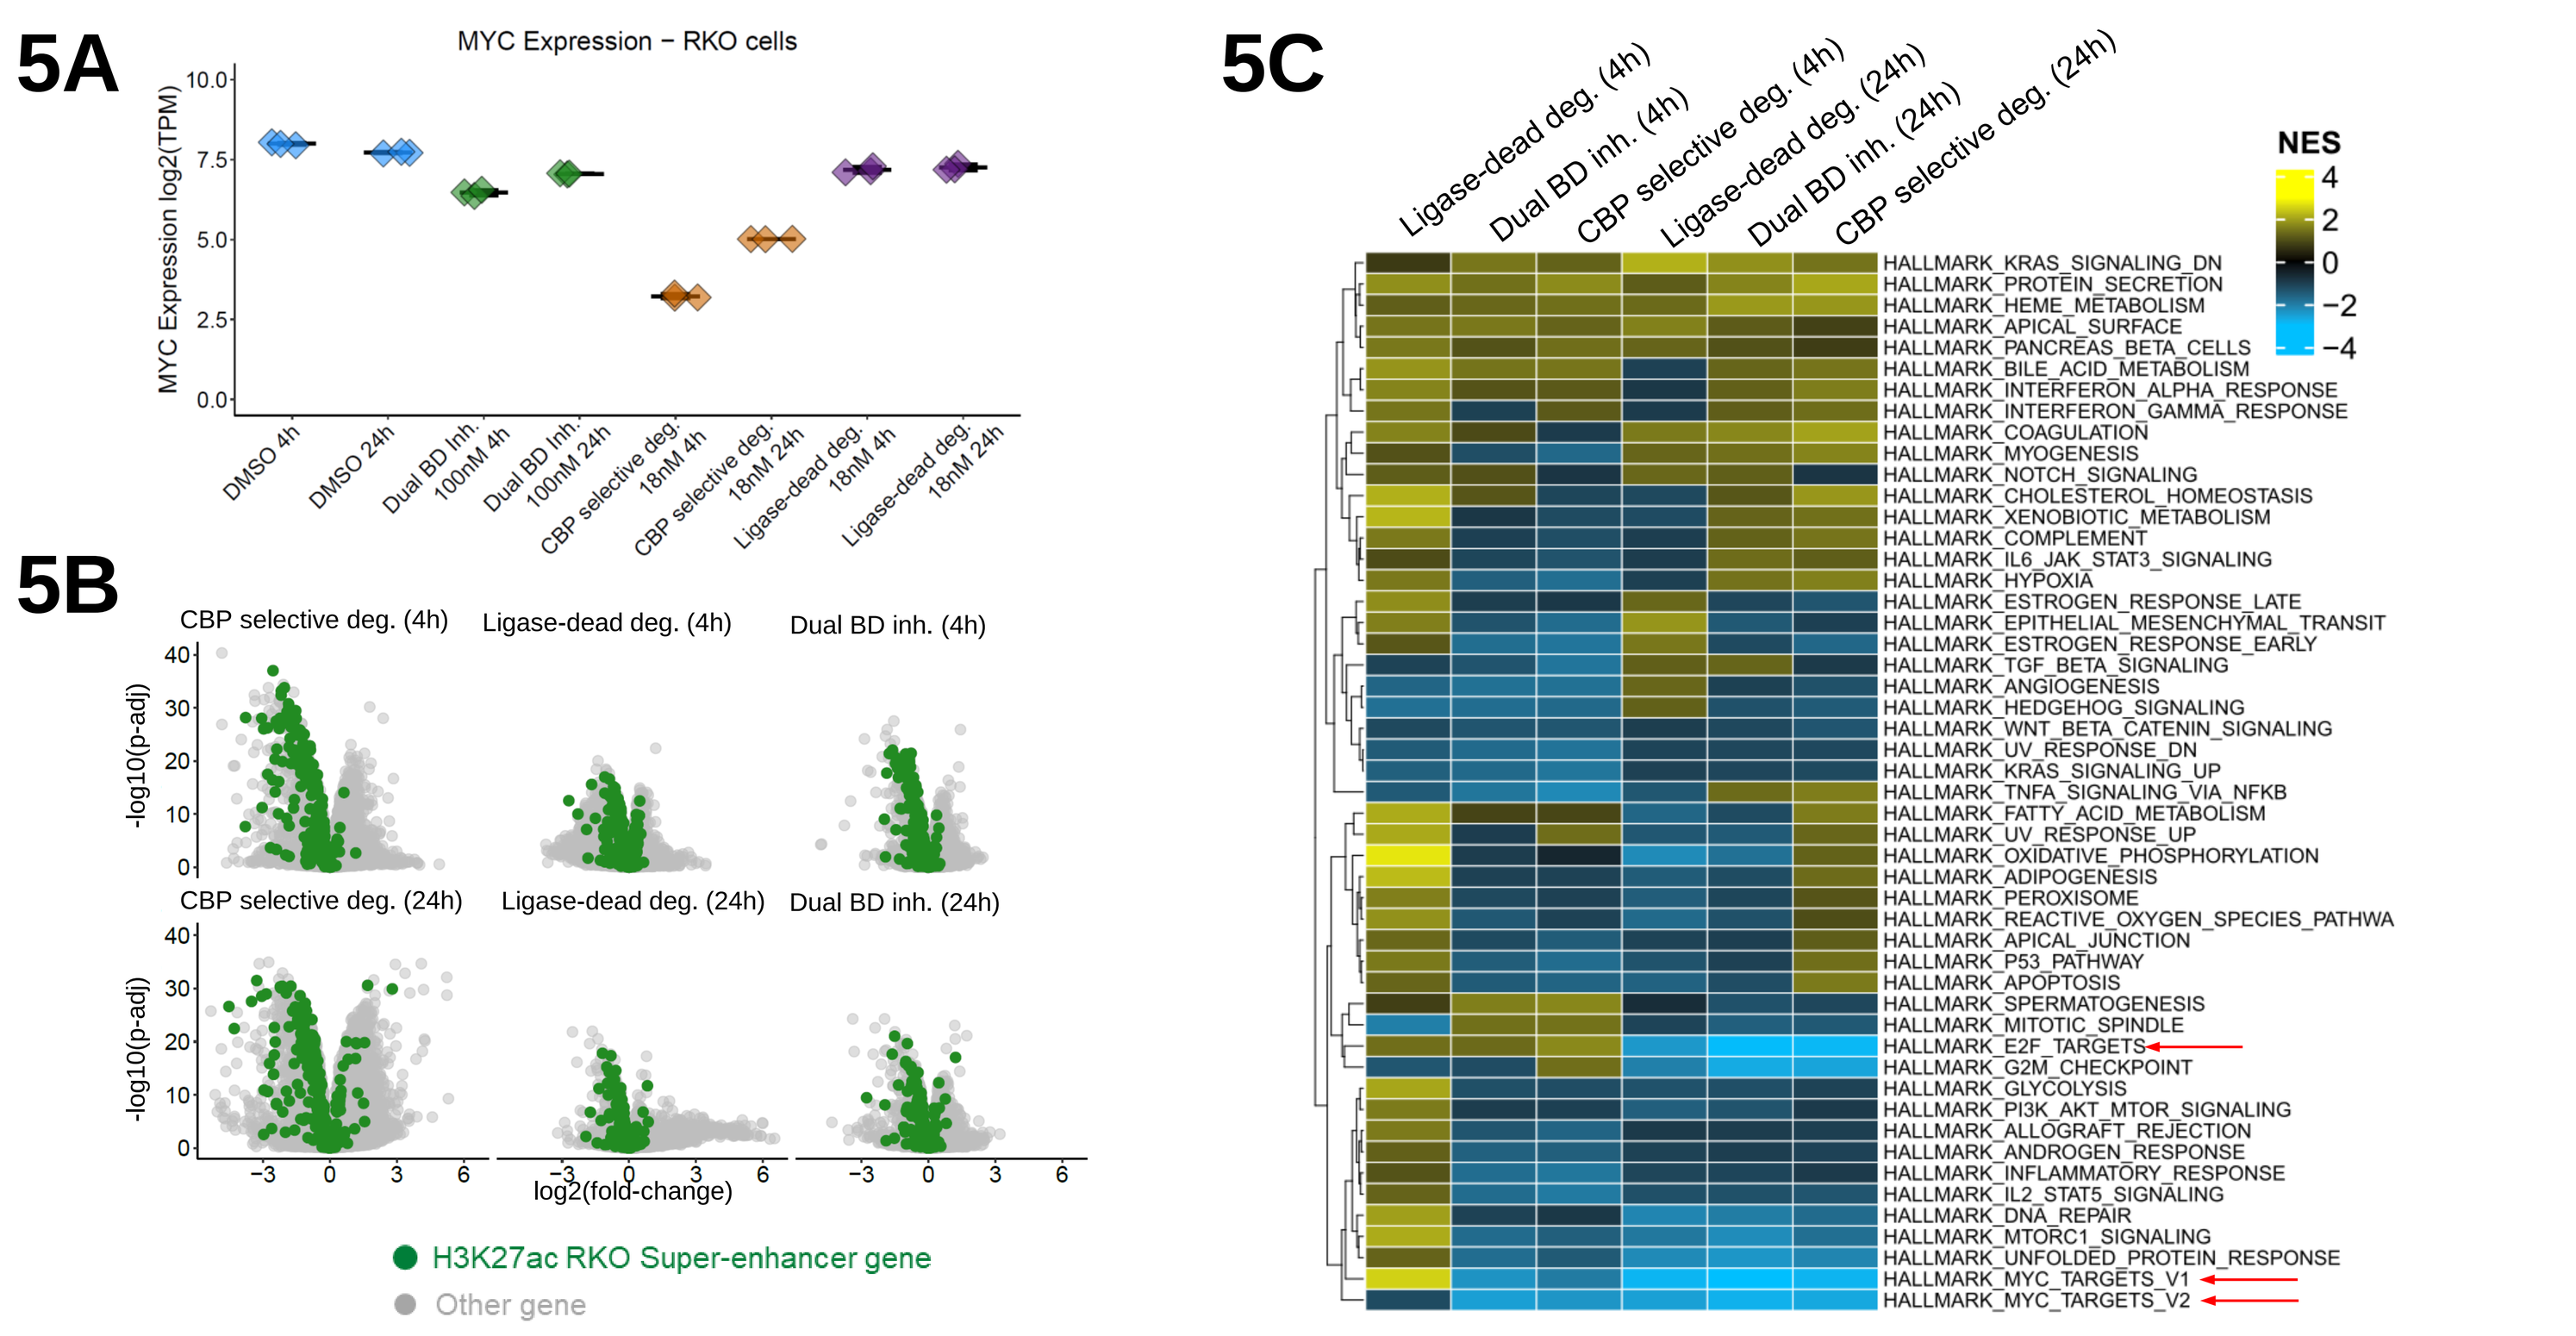
5A
MYC Expression – RKO cells
5B
H3K27ac RKO Super-enhancer gene
Other gene
5C
NES

Figure 5: RNA-seq analysis shows down-regulation of (A) c-Myc and (B) RKO super-enhancer genes (defined by H3K27ac) after CBP degradation C) GSEA shows MYC and E2F targets down-regulated at 24 hours after CBP degradation.

Figure 6. Thrombocytopenia focused safety studies

6A

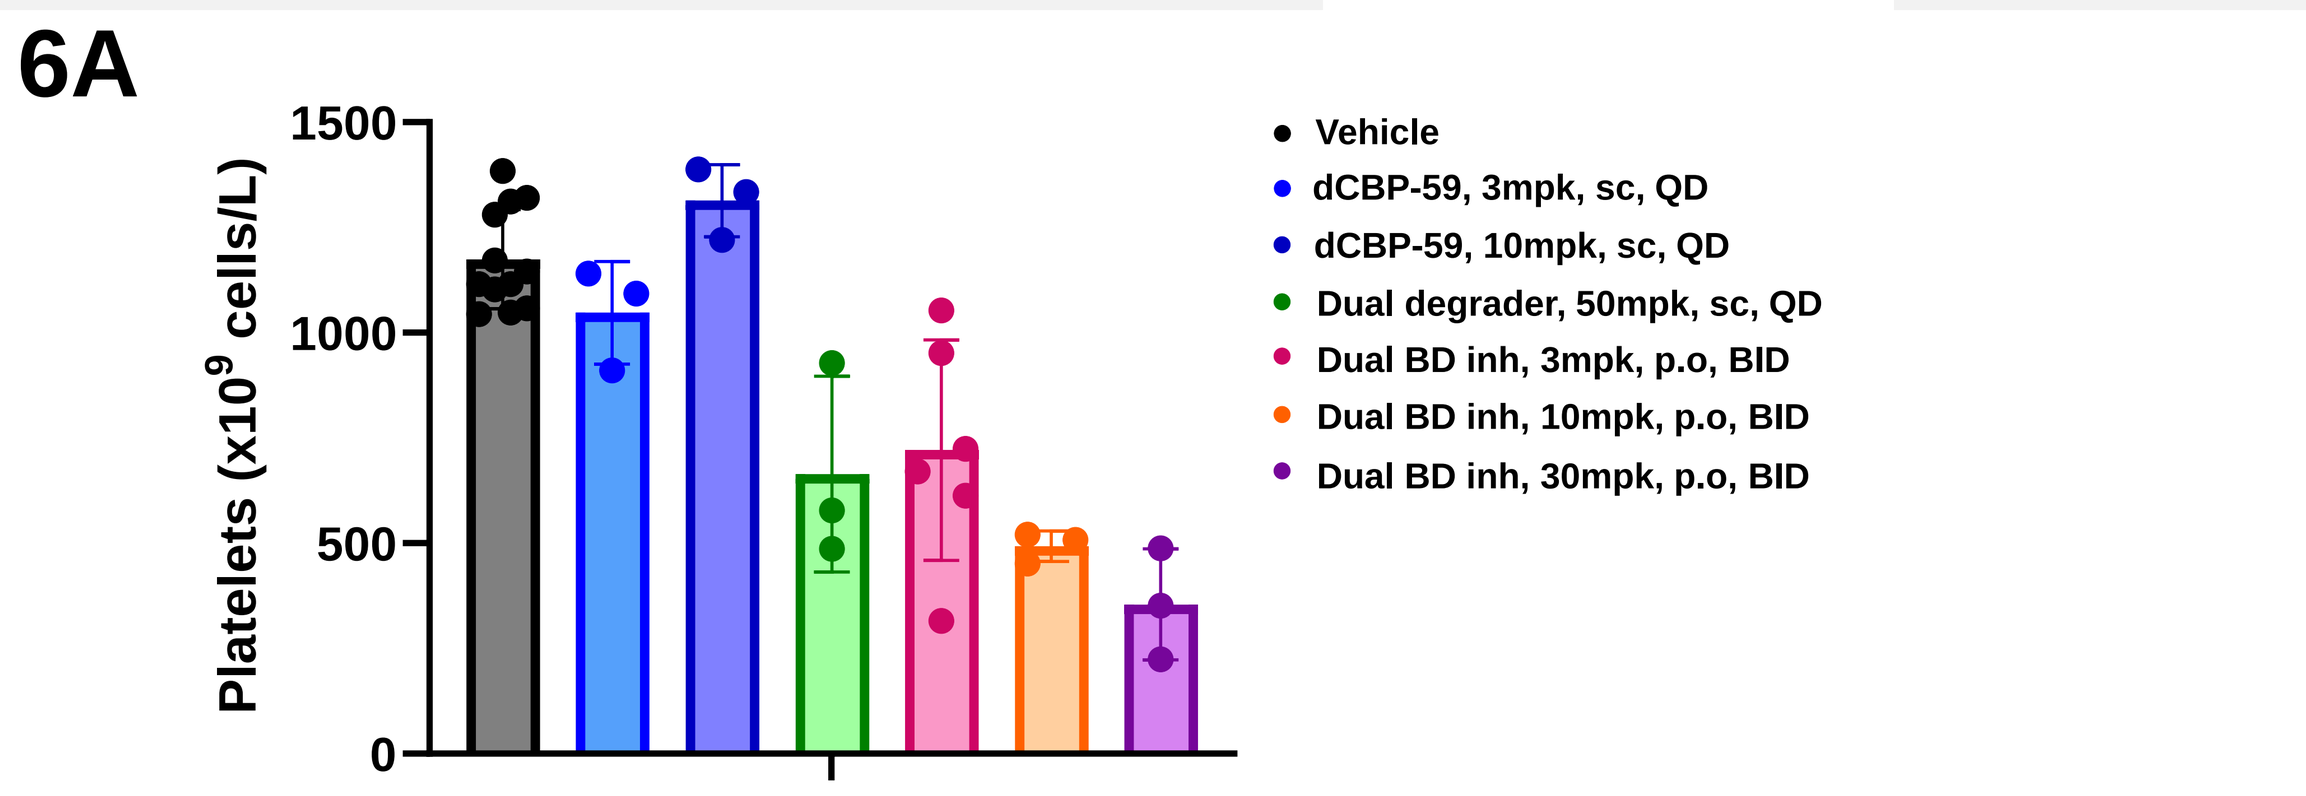
Platelets ($x10^9$ cells/L)
Vehicle
dCBP-59, 3mpk, sc, QD
dCBP-59, 10mpk, sc, QD
Dual degrader, 50mpk, sc, QD
Dual BD inh, 3mpk, p.o, BID
Dual BD inh, 10mpk, p.o, BID
Dual BD inh, 30mpk, p.o, BID

Figure 6A: Selective CBP degrader has no impact on total platelet counts compared to dual bromodomain inhibition following a 14 day mouse in vivo study.

6B

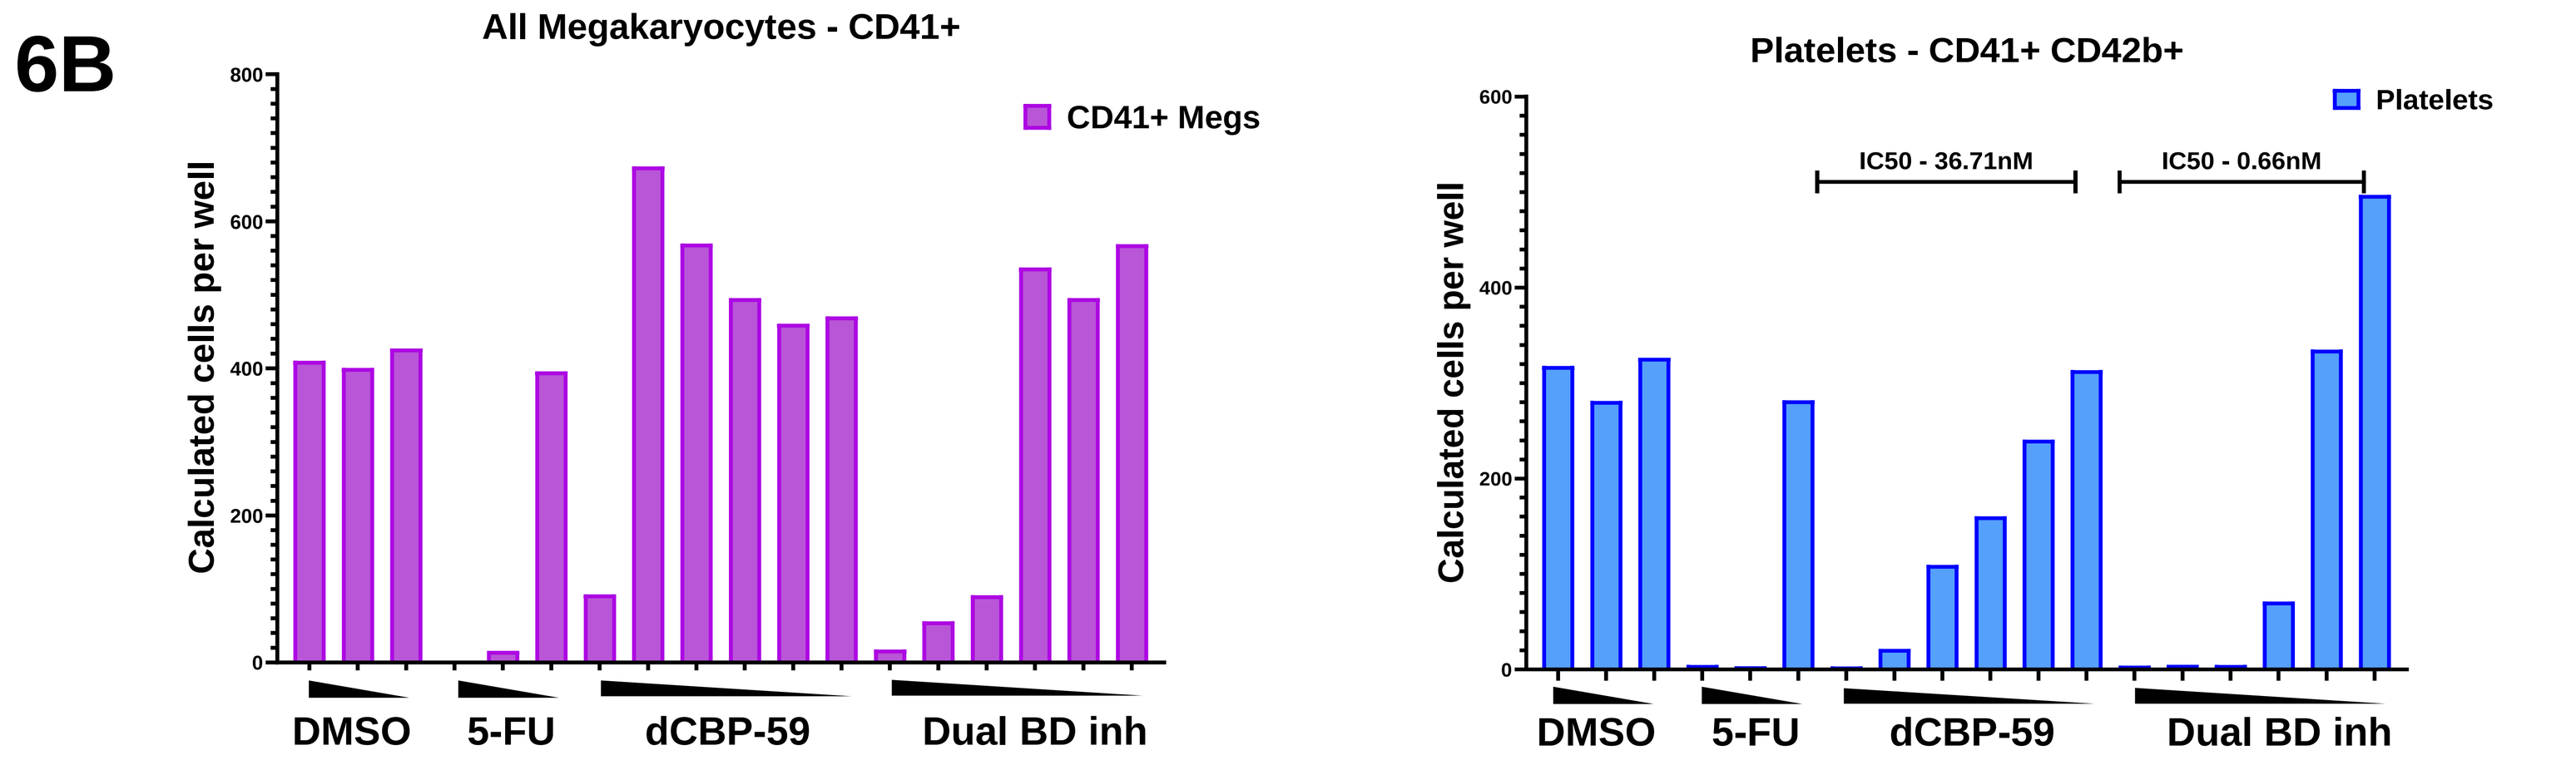
All Megakaryocytes - CD41+
CD41+ Megs
Platelets - CD41+ CD42b+
Platelets
IC50 - 36.71nM
IC50 - 0.66nM
Calculated cells per well
DMSO
5-FU
dCBP-59
Dual BD inh

Figure 6B: In vitro megakaryocyte differentiation and platelet measurement assay confirms increased safety with a selective degrader over dual bromodomain inhibition.

CMPD	EP300 Bromo Binding [uM]	CBP Bromo Binding [uM]
Dual BD inh	0.001	0.001
dCBP-59	0.050	0.008

CONCLUSIONS

- We have generated potent and selective CBP degraders that validate the synthetic lethal relationship and show anti-tumor activity across various EP300 mutant cell lines from multiple indications.
- Using our in vivo enabled compounds we have achieved efficacy in a number of different EP300 mutant xenograft models, across multiple indications of interest.
- We have identified and are currently expanding our understanding of the mechanism of action and biomarkers of selective CBP degradation.
- We show that selective degradation of CBP results in a lower risk of thrombocytopenia as compared to dual CBP/EP300 bromodomain inhibitors.

ACKNOWLEDGEMENTS: The team owes many thanks to Ammar Adam, GiNell Elliott, Janice Lee, Solymar Negretti, Matt Netherton, Brenna Sherbanee, Lee Silverman and David Terry for their support and contributions.