Discovery and Characterization of Novel, Selective EP300 Degraders

Darshan Sappal, Laura La Bonte, Ammar Adam, Benjamin Adams, Ketaki Adhikari, Wesley Austin, Breanna Bullock, Danette Daniels, Julie Di Bernardo, Thomas Dixon, GiNell Elliot, Md Imran Hossain, Dave Lahr, Mei Yun Lin, Karolina Mizeracka, Solymar Negretti, Shawn Schiller, Brenna Sherbanee, David Terry, Nihan Ucisik, Elizabeth Wittenborn, Mark Zimmerman

FOGHORN® THERAPEUTICS

INTRODUCTION

- CREB binding protein (CBP) and E1A binding protein (EP300) are paralog histone acetyltransferases involved in many cellular processes via their activity as transcription factor co-activators
- Dysregulation has been implicated across a variety of cancer indications
- Due to the high homology between EP300 and CBP, identification of selective chemical matter that specifically and directly targets EP300 has proven challenging.
- Herein we describe a potent, highly selective EP300 heterobifunctional degrader with selective biological activity in EP300 dependent cancer cell lines.

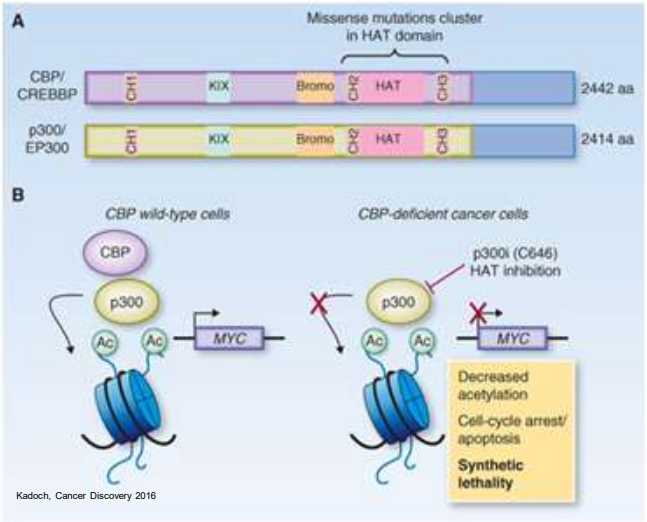
A
Missense mutations cluster in HAT domain
CBP/ CREBBP
p300/ EP300
CH1
KIX
Bromo
CH2
HAT
CH3
2442 aa
2414 aa
B
CBP wild-type cells
CBP-deficient cancer cells
CBP
p300
Ac
MYC
p300i (C646) HAT inhibition
Decreased acetylation
Cell-cycle arrest/ apoptosis
Synthetic lethality
Kadoch, Cancer Discovery 2016

Figure 1. An EP300 selective therapy has the potential to address over 100,000 patients per year across many indications

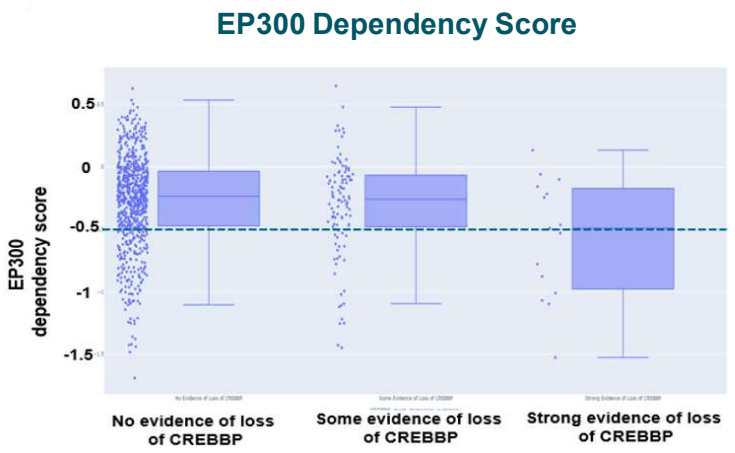
EP300 Dependency Score
EP300 dependency score
0.5
0
-0.5
-1
-1.5
No evidence of loss of CREBBP
Some evidence of loss of CREBBP
Strong evidence of loss of CREBBP

Target / Approach	· E1A binding protein p300 (EP300) · Targeted protein degrader
Initial Indication	· CBP mutated and subsets of EP300 dependent hematologic malignancies (e.g., Bladder, Prostate, Lymphoma, Myeloma, T-ALL, DLBCL, AML, CML)
Mutation / Aberration	· CBP mutated cancers · EP300 dependent cancers
Stage	· Pre-clinical
New Patients Impacted / Year*	· Over 100,000

* U.S., EU5, Japan.

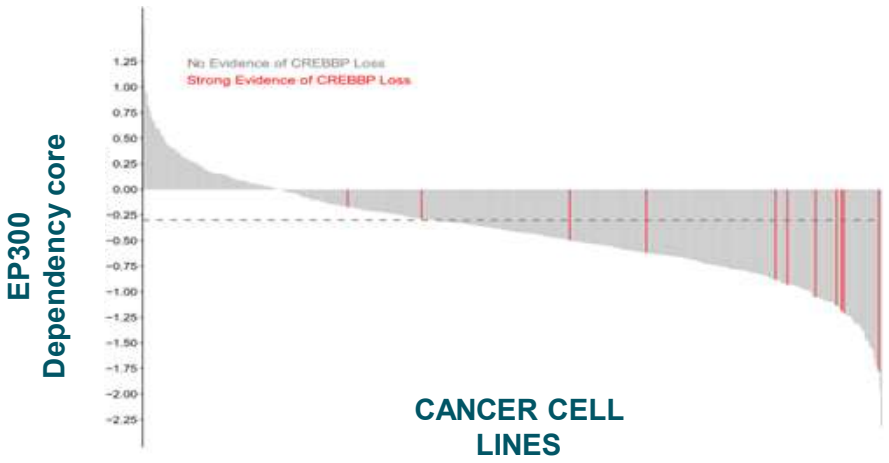
EP300 Dependency core
No Evidence of CREBBP Loss
Strong Evidence of CREBBP Loss
CANCER CELL LINES

Figure 1: The DepMap Portal was used to identify cell lines with strong dependency on EP300 either based on the bi-directional synthetic lethal relationship with CBP or based on general dependency on EP300. This criteria for cell line selection was then applied to patient tumor databases to extrapolate treatable patient populations

RESULTS

Figure 2. Identification of potent, selective EP300 Degraders

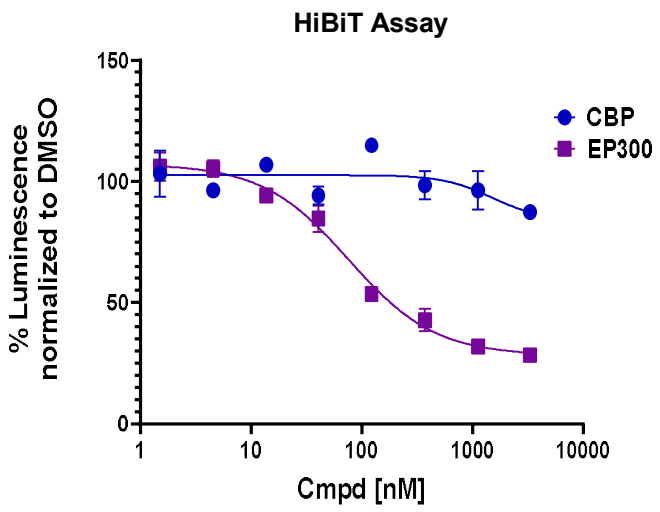
HiBiT Assay
% Luminescence normalized to DMSO
CBP
EP300
Cmpd [nM]

In Vitro Properties	Compound 1
Cellular Potency DC_{50} (KI HiBiT)	0.069 µM
Dmax (KI HiBiT)	74%
LogD	3.77
FaSSIF	13.1 µM

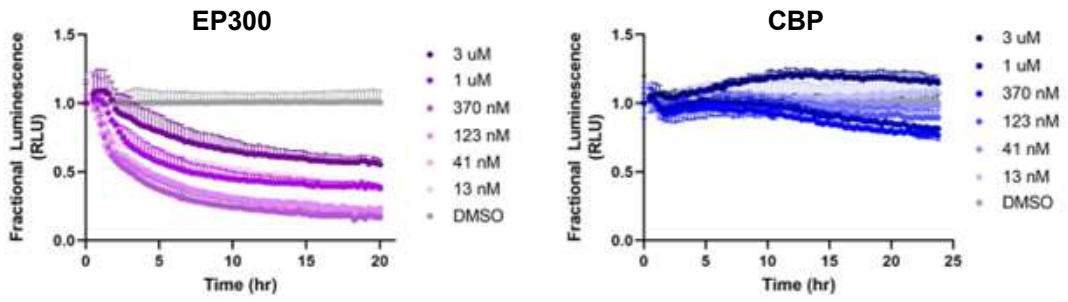
EP300
CBP
Fractional Luminescence (RLU)
Time (hr)
3 uM
1 uM
370 nM
123 nM
41 nM
13 nM
DMSO

Figure 2: We have identified potent and selective degraders of EP300. Dose response kinetic data confirm EP300 selectivity.

Figure 3a. EP300 selective degraders show an antiproliferative effect in CBPmut and EP300 dependent cell lines, but not wild type

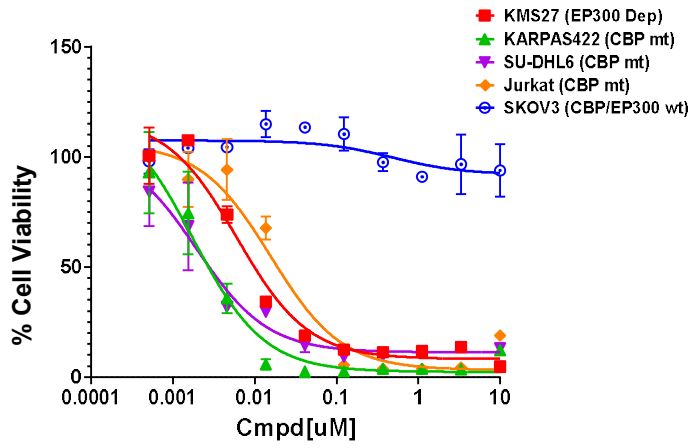
KMS27 (EP300 Dep)
KARPAS422 (CBP mt)
SU-DHL6 (CBP mt)
Jurkat (CBP mt)
SKOV3 (CBP/EP300 wt)
% Cell Viability
Cmpd[uM]

Figure 3b. EP300 selective degraders demonstrate antiproliferative effects in AR+ prostate cell lines

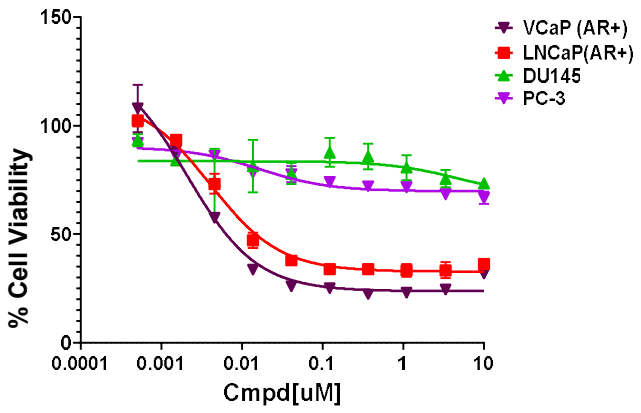
VCaP (AR+)
LNCaP(AR+)
DU145
PC-3
% Cell Viability
Cmpd[uM]

Figure 3a, 3b: EP300 degraders exhibit potent antiproliferative effects in EP300 dependent cell lines (including CBP mt lines) (Fig3a). In particular, we observe greater sensitivity in AR+ vs. AR- prostate cancer cell lines (Fig 3b).

Figure 4. EP300 selective degraders demonstrate robust degradation in vivo

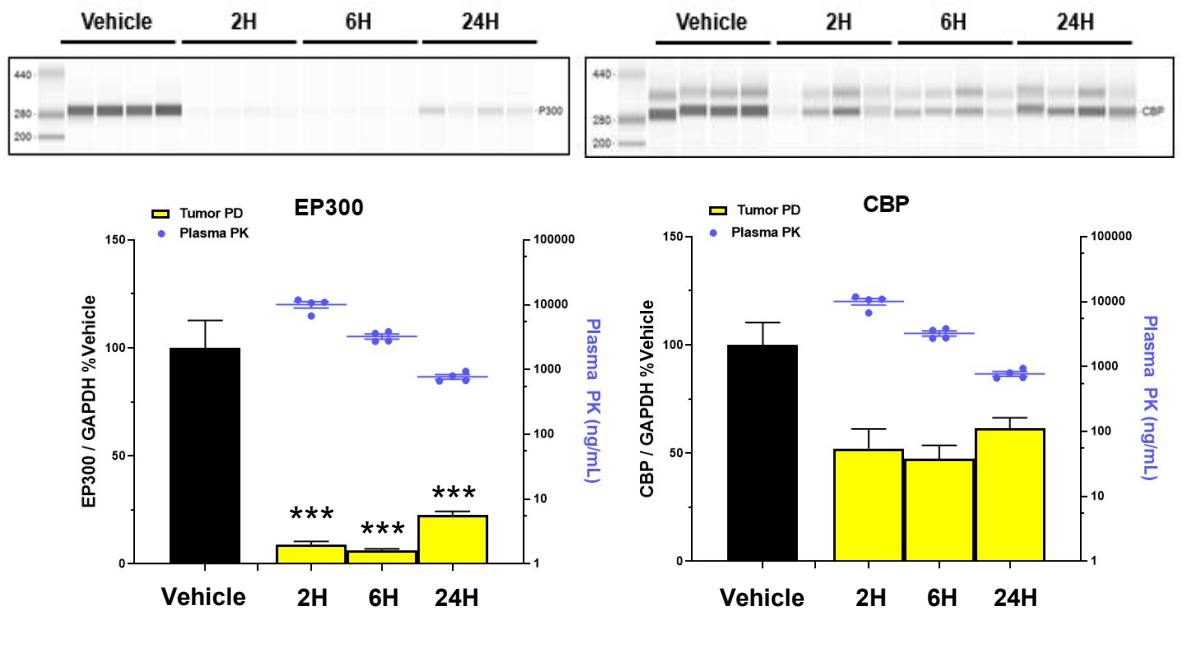
Vehicle
2H
6H
24H
EP300
CBP
Tumor PD
Plasma PK
EP300 / GAPDH %Vehicle
CBP / GAPDH %Vehicle
Plasma PK (ng/mL)

***p<0.0001 compared w/vehicle by One-way ANOVA

Figure 4: EP300 selective degraders show robust, sustained EP300 degradation in vivo

CONCLUSIONS

- EP300 and its paralog CBP are transcriptional co-activators that have been associated with several oncogenic signaling pathways.
- If successful, the identification and development of a selective EP300 degrader may address an unmet medical need for a significant patient population.
- Here, we describe potent, selective EP300 degraders that show anti-proliferative effects in CBP mutant and EP300 dependent cell lines while sparing wild type cell lines
- Our EP300 degraders show increased sensitivity in AR+ vs. AR- Prostate Cancer cell lines.
- Additionally, our selective EP300 degraders are in vivo enabled, and demonstrate robust, selective degradation of EP300 in vivo.
- CBP and EP300 share many overlapping but also non-overlapping functions. Our selective tool degraders will also help us understand and identify cancer indications where the dependency on one of the targets might be agnostic of the paralog’s mutational status.

ACKNOWLEDGEMENTS: Thank you to Janice Lee, Matt Netherton and Ricardo Medina for their consulting role in the program

REFERENCES: Kadoch C. Lifting up the HAT: Synthetic lethal screening reveals a novel vulnerability at the CBP-EP300 axis. Cancer Discov. 2016
Sources for incidence numbers: US - SEER Database, EU - ECIS Database, JP - WHO Globocan, Foghorn TCGA and GENIE Analysis, DRG Reports