Characterizing Compound Behavior at Foghorn Therapeutics

Brenna Sherbanee[1], Julie Di Bernardo[1], Silvia Escudero[1], and Marina Nelen[1]

[1]Foghorn Therapeutics, Drug Discovery, Cambridge, MA

https://foghorntx.com/

Abstract

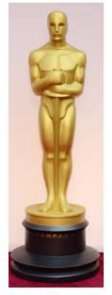

Bad actors in assays:
- Detrimental for instruments (SPR, LC/MS..)
- Assay interference

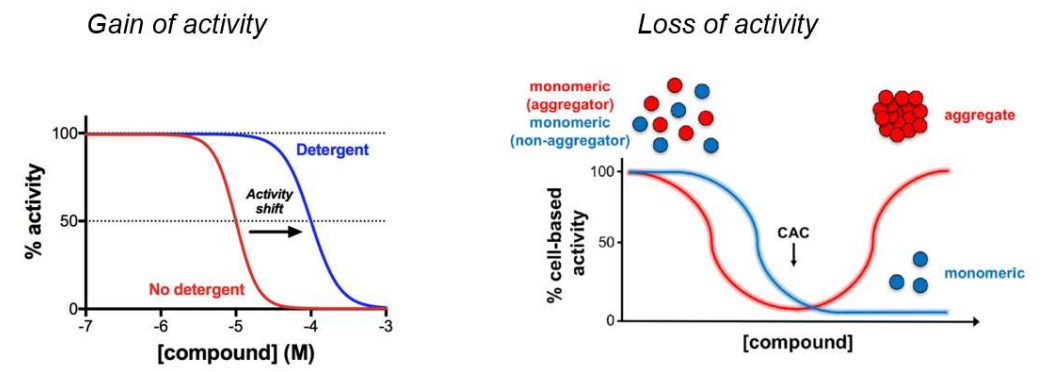
Gain of activity
Loss of activity
% activity
Detergent
Activity shift
No detergent
[compound] (M)
monomeric (aggregator)
monomeric (non-aggregator)
aggregate
% cell-based activity
CAC
monomeric
[compound]

Fig 1. Not all compounds are created equal: the earlier we gain insights in compound behavior in solution, the faster we can make informed decisions about following up on a hit or de-prioritize it. Bad actors are known to damage instruments (precipitation on SPR chips, clogging of columns, etc…) and they often confuse assay results (false positive and false negative hits) [1].

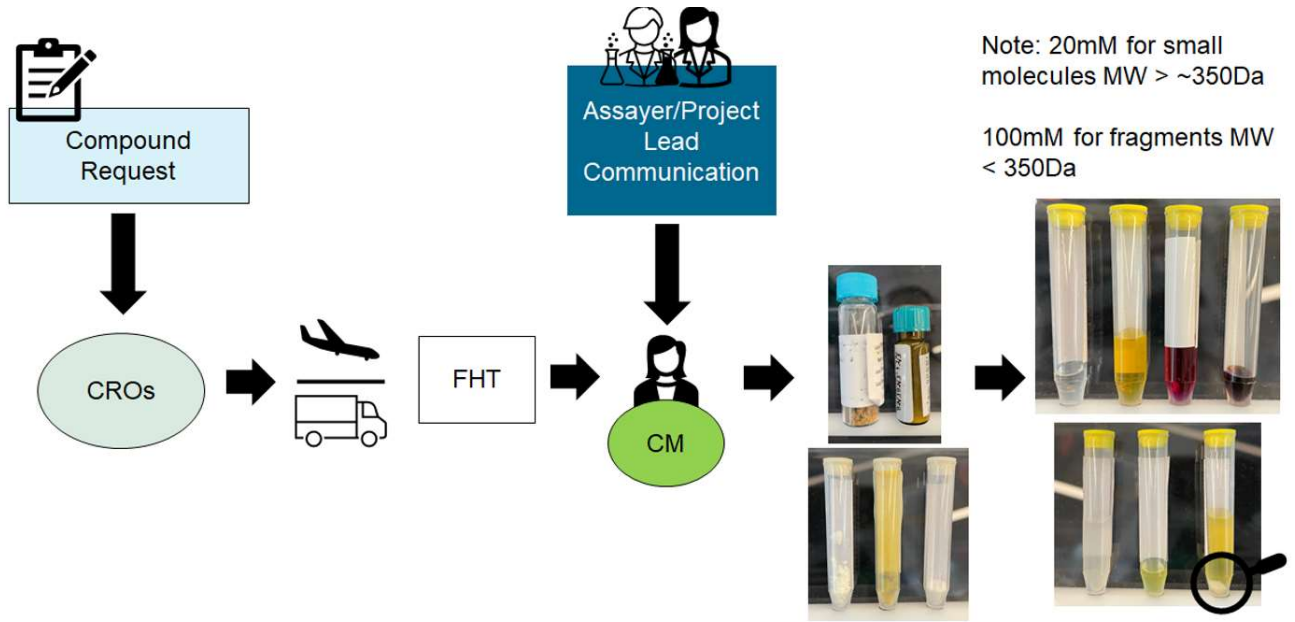
Compound Request
Assayer/Project Lead Communication
Note: 20mM for small molecules MW > ~350Da
100mM for fragments MW < 350Da
CROs
FHT
CM

Fig 2. Compounds visual inspection: schematic of how Compound Management is the first source of information about compound appearance as solid and behavior in DMSO.

Assays in our toolkit

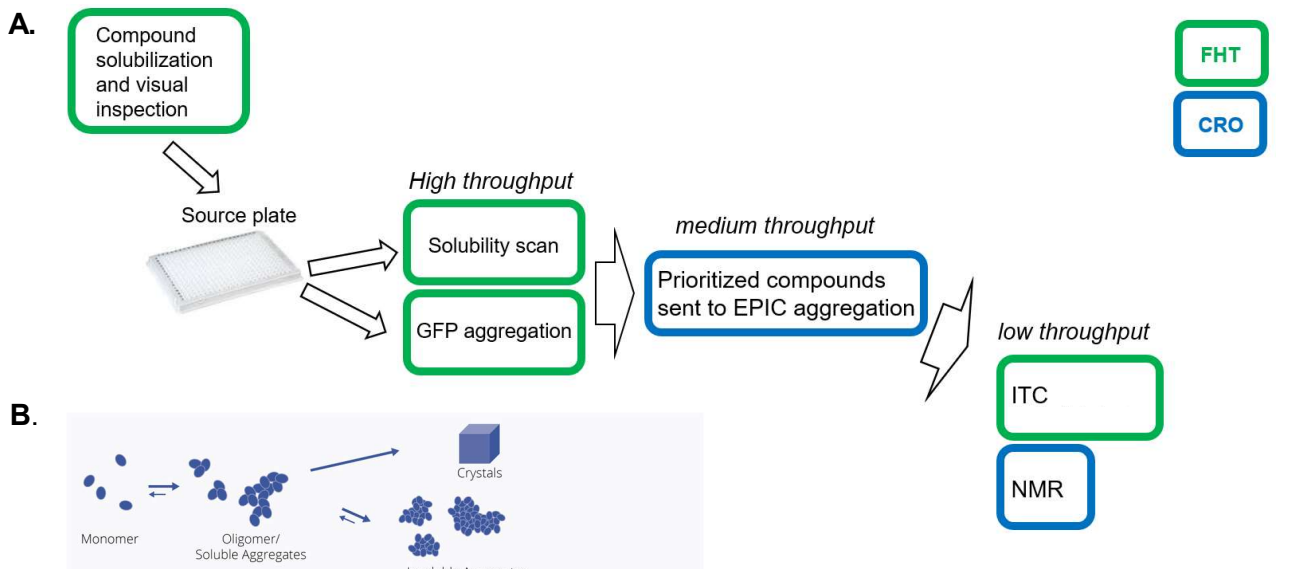
A.
Compound solubilization and visual inspection
Source plate
High throughput
Solubility scan
GFP aggregation
medium throughput
Prioritized compounds sent to EPIC aggregation
low throughput
ITC
NMR
FHT
CRO

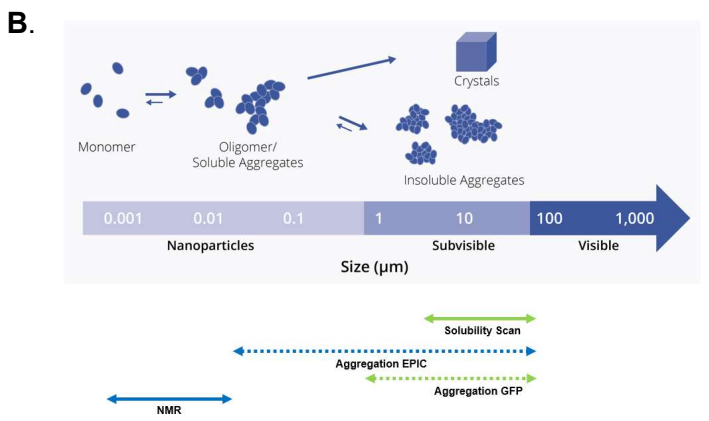
B.
Crystals
Monomer
Oligomer/ Soluble Aggregates
Insoluble Aggregates
0.001
0.01
0.1
1
10
100
1,000
Nanoparticles
Subvisible
Visible
Size (µm)
Solubility Scan
Aggregation EPIC
Aggregation GFP
NMR

Fig 3. FHT assay toolkit for compound profiling: A. Schematic of assays based on their throughput. **B.** Schematic of what the assays are capturing in terms or aggregate size [2].

1. Solubility scan

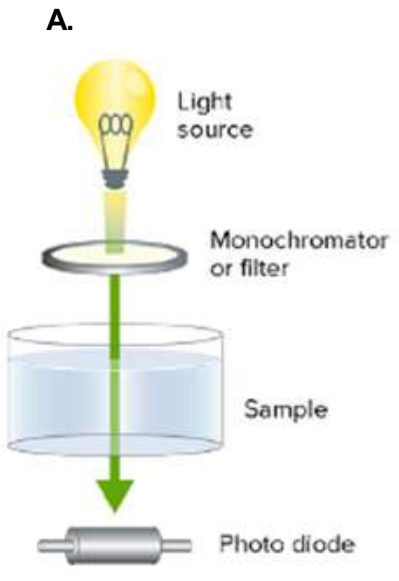
A.
Light source
Monochromator or filter
Sample
Photo diode

Fig 4. Solubility scan: Compound and buffer are mixed, incubated, then absorbance is read at 600 nm. DMSO is used as the control, and any precipitation will result in an increase in absorbance. If these absorbance values fall outside the standard deviation of the DMSO controls, these values "fail", and the compound has a lower solubility limit. (Ex. – as seen in **B.** – FHT-002 has a solubility limit of 87.8 uM, since absorbance values at higher concentrations fall outside the SD). **A**. Absorbance scan [3]. **B**. Example of solubility data analysis.

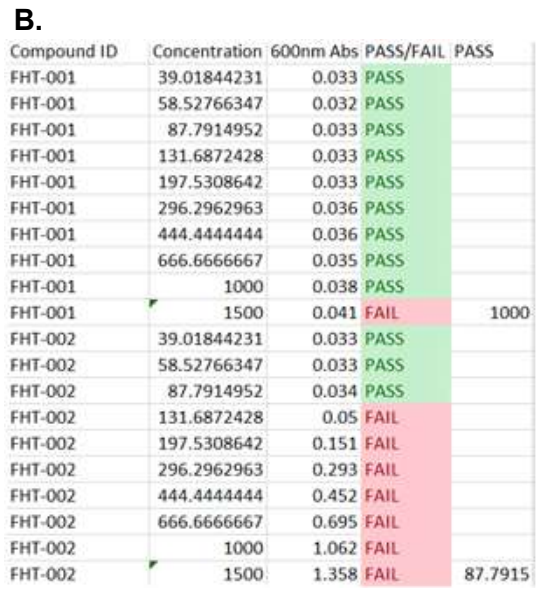

B.

Compound ID	Concentration	600nm Abs	PASS/FAIL	PASS
FHT-001	39.01844231	0.033	PASS	
FHT-001	58.52766347	0.032	PASS	
FHT-001	87.7914952	0.033	PASS	
FHT-001	131.6872428	0.033	PASS	
FHT-001	197.5308642	0.033	PASS	
FHT-001	296.2962963	0.036	PASS	
FHT-001	444.4444444	0.036	PASS	
FHT-001	666.6666667	0.035	PASS	
FHT-001	1000	0.038	PASS	
FHT-001	1500	0.041	FAIL	1000
FHT-002	39.01844231	0.033	PASS	
FHT-002	58.52766347	0.033	PASS	
FHT-002	87.7914952	0.034	PASS	
FHT-002	131.6872428	0.05	FAIL	
FHT-002	197.5308642	0.151	FAIL	
FHT-002	296.2962963	0.293	FAIL	
FHT-002	444.4444444	0.452	FAIL	
FHT-002	666.6666667	0.695	FAIL	
FHT-002	1000	1.062	FAIL	
FHT-002	1500	1.358	FAIL	87.7915

2. GFP aggregation assay

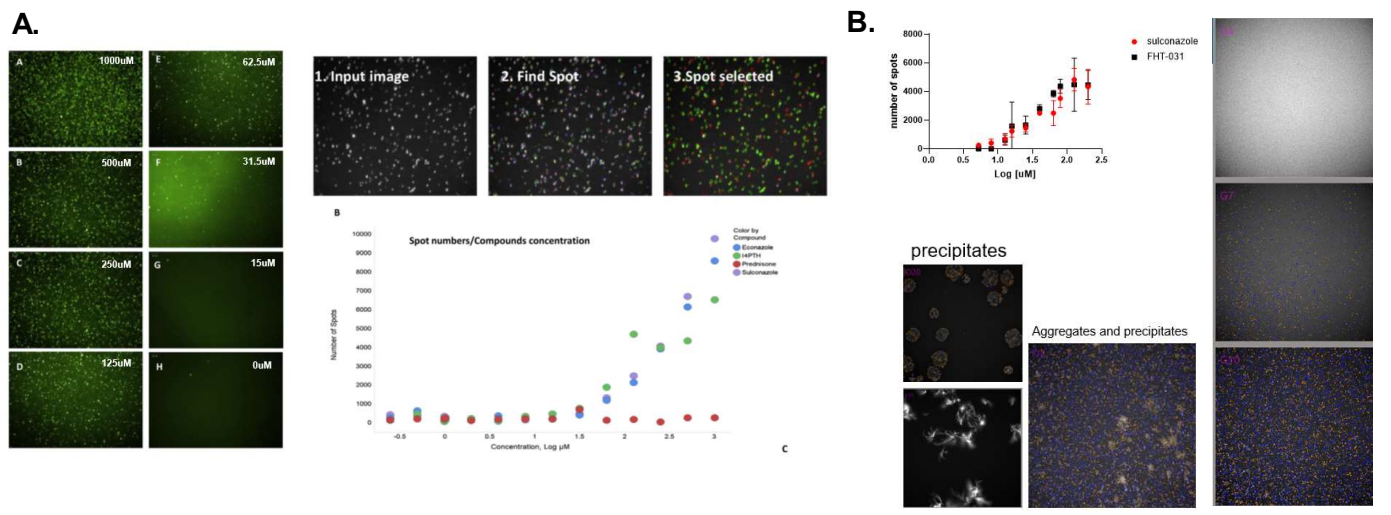
A.
1000uM
62.5uM
500uM
31.5uM
250uM
15uM
125uM
0uM
1. Input image
2. Find Spot
3.Spot selected
Spot numbers/Compounds concentration
B.
sulconazole
FHT-031
Log [uM]
precipitates
Aggregates and precipitates

Fig 5. Aggregation by imaging: GFP protein and compounds are mixed in buffer. If the compound is 'clean' and doesn't aggregate, GFP is disperse in solution and fluoresces in a very homogeneous way (DMSO marked as 0uM in **A.** and DMSO marked as G4 in **B.**). If the compound is an aggregator, it incapsulates GFP causing the protein to focalize in bright puncta. The more the compound aggregates, the more puncta can be detected. Imager software will then quantify the puncta and exclude any other auto-fluorescent artifacts such as large precipitated crystals (as seen in **B.**, labeled as 'precipitates'). **A.** literature data [4]; **B.** in-house examples.

3. EPIC aggregation assay

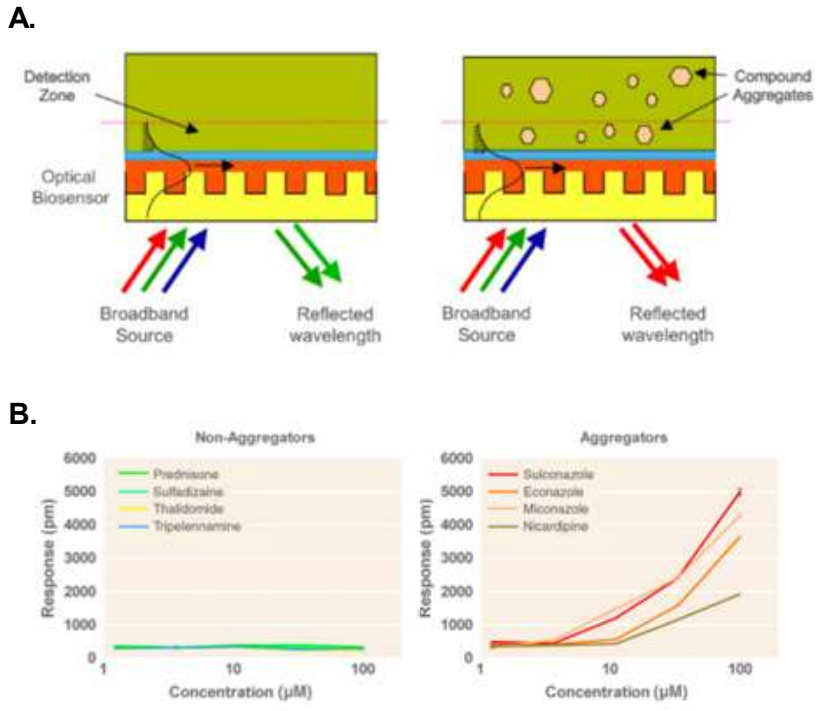
A.
Detection Zone
Optical Biosensor
Broadband Source
Reflected wavelength
Compound Aggregates
B.
Non-Aggregators
Prednisone
Sulfadiazine
Thalidomide
Tripelennamine
Aggregators
Sulconazole
Econazole
Miconazole
Nicardipine
Response (pm)
Concentration (µM)

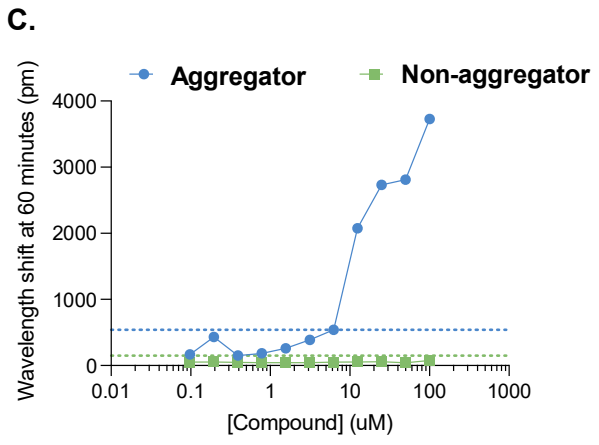
C.
Aggregator
Non-aggregator
Wavelength shift at 60 minutes (pm)
[Compound] (uM)

Fig 6. Aggregation by EPIC: A. Schematic of EPIC aggregation assay. Presence of compound aggregates in solution cause a shift in the wavelength of light reflected off an optical biosensor at the bottom of the plate well [5]. **B.** Plotting the shift in wavelength versus compound concentration results in aggregation curves depicting the concentration at which soluble aggregates begin to form. [4]. **C.** Internal data collected at Biophysical Solutions Inc. showing the difference between an aggregator (blue) and non-aggregator (green). Dotted lines represent the assay baseline for each compound.

4. NMR

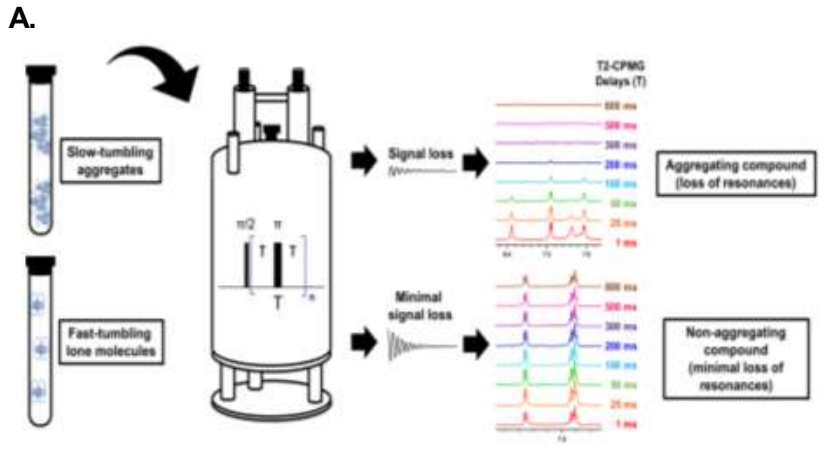
A.
Slow-tumbling aggregates
Fast-tumbling lone molecules
Signal loss
Minimal signal loss
T2-CPMG Delays (T)
Aggregating compound (loss of resonances)
Non-aggregating compound (minimal loss of resonances)

Fig 7. Aggregation by NMR: A. Schematic of NMR aggregation assay using T2-CPMG method. Compound aggregates cause a loss of signal as the T2 relaxation time increases due to slow tumbling of the larger species [6]. **B.** Internal data collected at NMX showing the difference between an aggregator (right) and a non-aggregator (left).

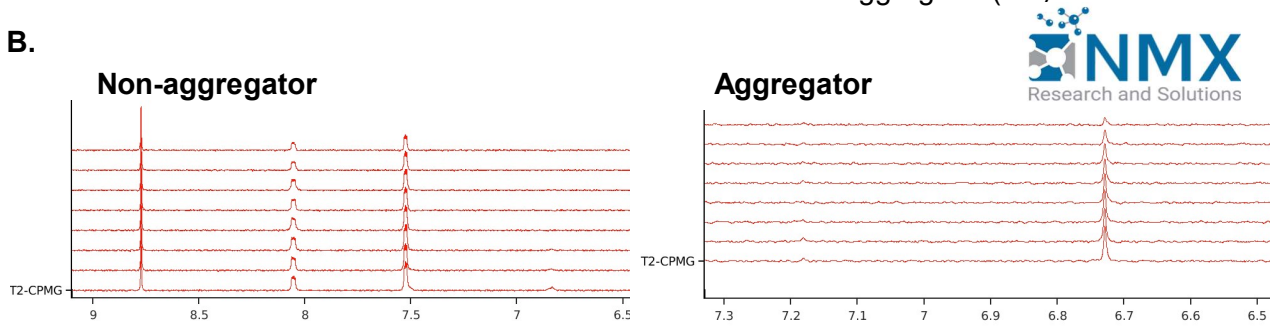
B.
Non-aggregator
Aggregator
NMX
Research and Solutions
T2-CPMG

Assay comparison

Compound	Solubility Limit (uM)	EPIC Turbidity Limit (uM)	GFP Assay Aggregator	NMR Aggregator
FHT-003	100	100	No	Possible
FHT-004	100	100	No	No
FHT-005	100	100	No	No
FHT-006	100	100	No	No
FHT-007	100	75	No	Yes
FHT-008	100	75	No	No
FHT-009	100	56	No	No
FHT-010	100	1	No	Possible
FHT-011	100	75	Possible	Possible
FHT-012	50	100	No	Possible
FHT-013	50	100	Possible	Possible
FHT-014	50	59	Possible	Yes
FHT-015	50	9	Possible	Possible
FHT-016	50	4	Possible	Possible
FHT-017	50	2	Possible	Possible
FHT-018	50	1	Possible	Yes
FHT-019	50	27.5	Yes	Yes
FHT-020	50	18	Yes	Possible
FHT-021	50	9	Yes	Possible
FHT-022	50	0.1	Yes	Possible
FHT-023	25	100	No	Yes
FHT-024	25	2.65	No	Yes
FHT-025	25	0.1	Possible	Possible
FHT-026	25	2	Yes	Possible
FHT-027	25	2	Yes	Possible
FHT-028	12.5	5	No	Possible
FHT-029	12.5	100	Possible	Possible
FHT-030	12.5	3	Yes	Possible

- Solubility and EPIC data generally correlate well **BUT:**
 - Good solubility but poor EPIC → Soluble aggregator
 - Poor solubility but good EPIC → Compound crashed out before reaching assay plate
- NMR is most sensitive technique but least high throughput and most expensive.
- Solubility, EPIC, and GFP data together filter out majority of poor-behaving compounds.

Fig 8. Compound behavior analysis across four assays.

Conclusions

No single assay in isolation can give a complete and definitive answer on compound behavior in solution. By running compounds through a combination of the various techniques, we can better assess the behavior/misbehavior of compounds and prioritize hits for follow-up.

References

[1]https://www.ncbi.nlm.nih.gov/books/NBK442297/#:~:text=Aggregation%20is%20a%20common%20mechanism,biochemical%20and%20cell%2Dbased%20assays.
[2]https://www.halolabs.com/applications/small-molecules-developing-better-drugs-through-the-unseen/
[3] https://www.moleculardevices.com/technology/absorbance
[4] Jiang et al., Development of a high-content imaging assay for screening compound aggregation, Analytical Biochemistry 559 (2018) 30-33
[5]https://www.corning.com/catalog/cls/documents/application-notes/snappshots_Label-Free_Detection_of_Compound_Aggregation_using_Corning_Epic_technology.pdf
[6]Ayotte et al. Exposing small-molecule nanoentities by a nuclear magnetic resonance relaxation assay, J. Med. Chem. 62 (2019) 7885-7896